EUROPEAN HEMATOLOGY ASSOCIATION

THE COMBINATION OF AXL INHIBITOR BEMCENTINIB AND LOW-DOSE CYTARABINE IS WELL TOLERATED AND EFFICACIOUS IN ELDERLY RELAPSED AML PATIENTS: UPDATE FROM THE ONGOING BGBC003 PHASE II TRIAL (NCT02488408)

BerGenBio

Sonja Loges*[1,2], Michael Heuser[3], Jörg Chromik[4], Carlos Enrique Vigil[5], Peter Paschka[6], Francesca Re[7], Nicola Di Renzo[8], Roberto Lemoli[9], Daniele Mattei[10], Isabel Ben Batalla[2,11], Monica Hellesøy[12], Jonas Waizenegger[2,11], Melanie Janning[2,11], Charles D Imbusch[2], Niklas Beumer[2], Austin Rayford[13], Jaya Neutiyal[14], Tzivia Berkman-Gottlieb[15], David Micklem[13], Hani Gabra[13], James B Lorens[16], Walter Fiedler[17], Yesid Alvarado-Valero[18], Bjørn T Gjertsen[19]

1 Personalized Oncology, University Hospital Mannheim, Mannheim, 2 German Cancer Research Center (DKFZ), Heidelberg, 3 Hematology, Hemostasis, Oncology and Stem Cell Transplantation, Hannover Medical School, Hannover , 4 University Hospital Frankfurt, Frankfurt, Germany, 5 Vanderbilt Ingram Cancer Centre, Nashville, United States, 6 University Hospital of Ulm, Ulm, Germany, 7 University of Parma, Parma, 8 Haematology and SCT Unit, Vito Fazzi Hospital, Lecce, 9 University of Genoa, Genoa, 10 ASO S. Croce e Carle, Cuneo, Italy, 11 Department of Personalised Oncology, University Medical Center Mannheim, Mannheim, Germany, 12 Haukeland University Hospital, 13 BerGenBio ASA, Bergen, Norway, 14 Translational Biomarkers, BerGenBio ASA, 15Data Analysis, BerGenBio , 16 Centre for Cancer Biomarkers, University of Bergen, Bergen, Norway, 17 University Medical Center Hamburg , Hamburg, Germany, 18 The University of Texas M.D. Anderson Cancer Center, Houston, United States, 19 Haukelands University Hospital, Bergen, Norway

Abstract EHA-2859

BACKGROUND, RATIONALE AND STUDY DESIGN

BGBC003 Study Rationale

- AXL, a receptor tyrosine kinase, is expressed in approx. 50% of all AML cases and is a negative prognostic factor[1]
- Bemcentinib is a first-in-class highly selective, potent, and orally bioavailable inhibitor of AXL
- Bemcentinib potentiates the efficacy of chemotherapy[2], prevents development of acquired drug resistance[3] and enhances the efficacy of immune checkpoint inhibitors[4] in several *in vivo* tumor models
- Bemcentinib inhibits survival of AML cells in vitro and enhances sensitivity to cytarabine in fresh AML blasts and AML cell lines[5]
- Bemcentinib is being developed as a monotherapy and in combination with chemotherapy in AML/MDS and a broader development program at BerGenBio is ongoing, including other indications

Proposed Mechanism

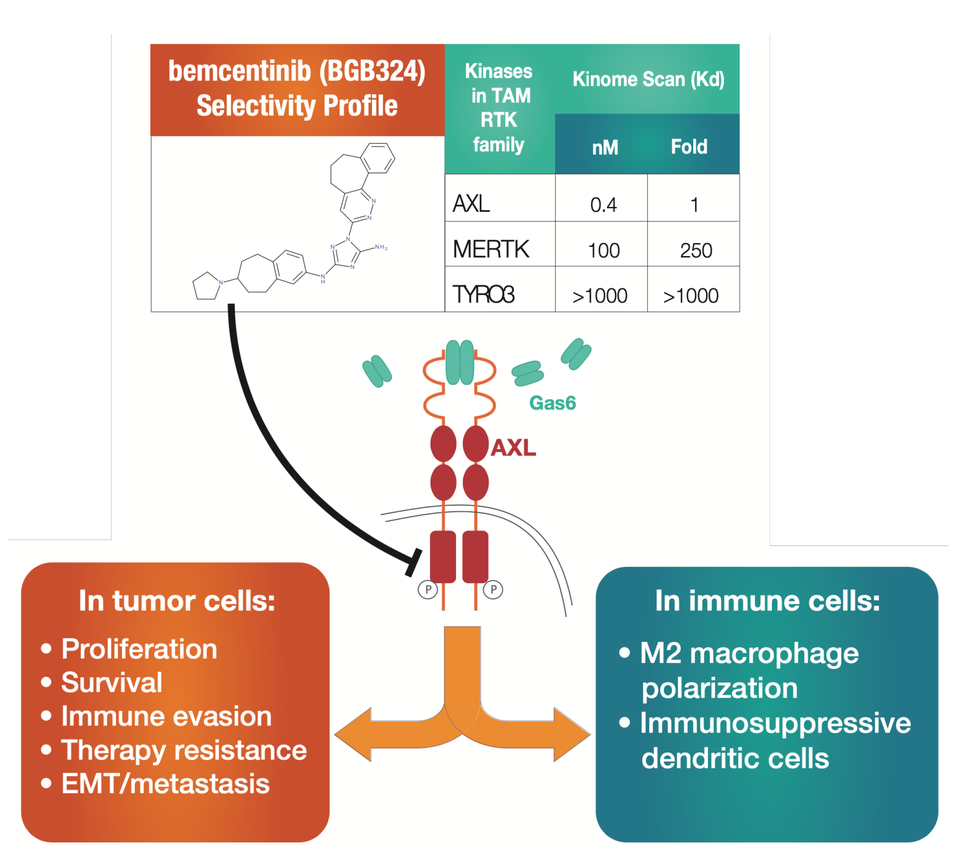

bemcentinib (BGB324) Selectivity Profile
Kinases in TAM RTK family
Kinome Scan (Kd)
nM
Fold
AXL 0.4 1
MERTK 100 250
TYRO3 >1000 >1000
Gas6
AXL
In tumor cells:
• Proliferation
• Survival
• Immune evasion
• Therapy resistance
• EMT/metastasis
In immune cells:
• M2 macrophage polarization
• Immunosuppressive dendritic cells

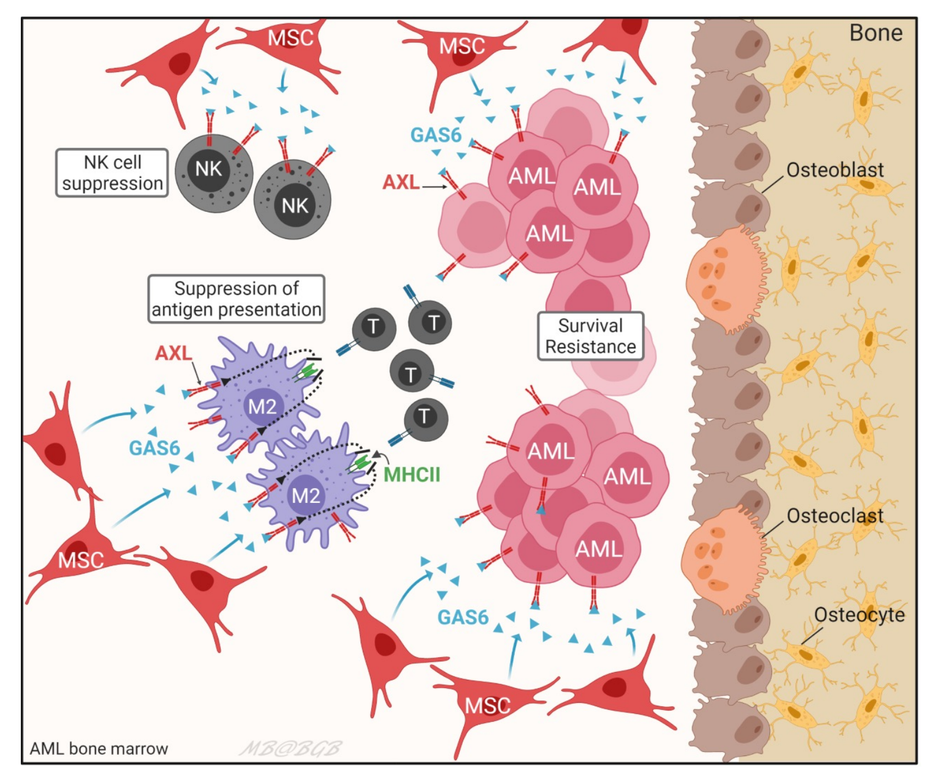

MSC
NK cell suppression
NK
GAS6
AXL
AML
Suppression of antigen presentation
T
M2
MHCII
Survival Resistance
Bone
Osteoblast
Osteoclast
Osteocyte
AML bone marrow

AML=acute myeloid leukemia; GAS6=growth arrest-specific protein 6; MSC=mesenchymal stem cell; M2= macrophage type 2; MHCII=major histocompatibility complex class II; NK=natural-killer; T=lymphocyte T

Study Design (with a focus on Bemcentinib+LDAC combination-cohorts B2 and B5)

Key inclusion criteria

- Diagnosis of AML (except FAB M3)
- Ineligible for intensive chemotherapy (advanced age or co-morbidities)
- Relapsed or refractory after at least 1 prior line of therapy
- Suitable to receive treatment with LDAC

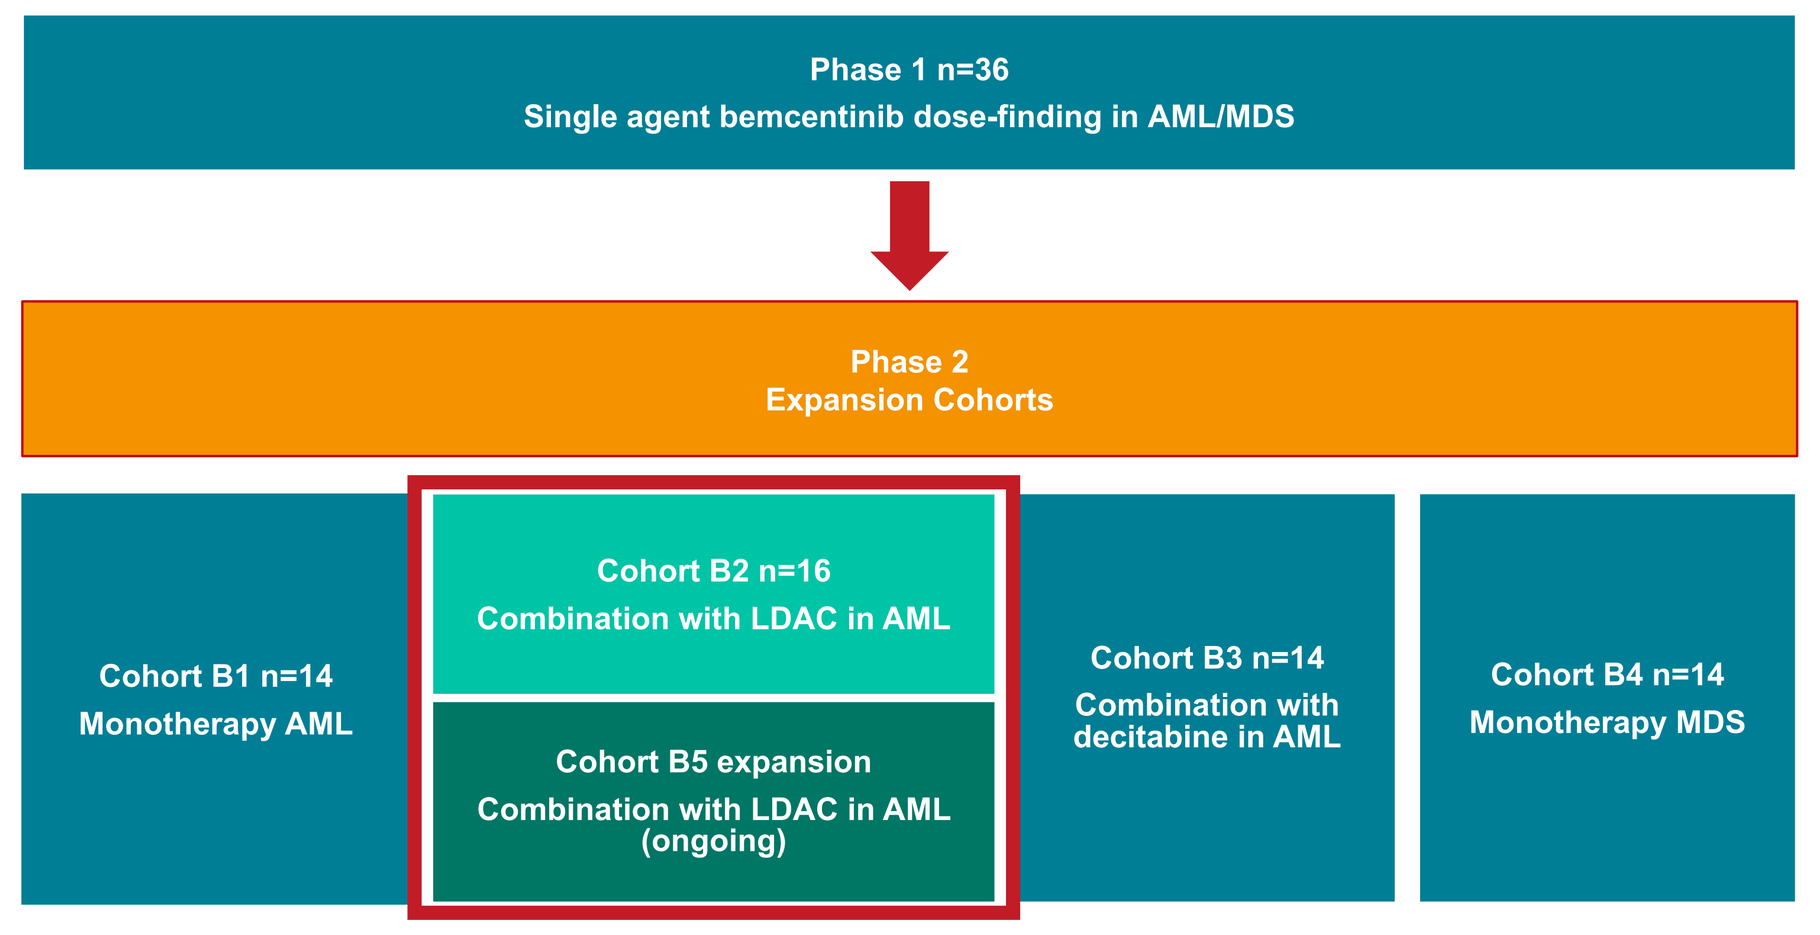

Phase 1 n=36
Single agent bemcentinib dose-finding in AML/MDS
Phase 2
Expansion Cohorts
Cohort B1 n=14
Monotherapy AML
Cohort B2 n=16
Combination with LDAC in AML
Cohort B5 expansion
Combination with LDAC in AML (ongoing)
Cohort B3 n=14
Combination with decitabine in AML
Cohort B4 n=14
Monotherapy MDS

LDAC = Low Dose Cytarabine; AML = Acute Myeloid Leukaemia; MDS = Myelodysplastic syndromes

Endpoints

Primary:
- Safety and tolerability

Secondary:
- ORR (overall response rate)
- RFS (relapse-free survival)
- OS (overall survival)
- PK profile

Demographics and disease characteristics

All patients in B2 and B5, n=34

Demographics and disease characteristics		n	%
Sex			
	Male	23	68%
	Female	11	32%
Age (years)			
	Median 76	(range 66-86)	
	<75 years	10	29%
	≥75 years	24	71%
ECOG at screening			
	0	11	32%
	1	18	53%
	2	4	12%
	Unknown	1	3%
% blasts at screening (bone marrow)			
	Median 38	(range 3-96)	
	<20%	7	21%
	≥20%	26	76%
	Unknown	1	3%
No. lines previous therapies			
	Median 1	(range 0-8)	
	0	7	21%
	1	13	38%
	2	8	24%
	≥3	6	18%
Disease status			
	Newly-diagnosed	7	21%
	Relapsed	20	59%
	Refractory	7	21%

Relapsed AML pts in B2 and B5, n=20

Demographics and disease characteristics		n	%
Sex			
	Male	12	60%
	Female	8	40%
Age (years)			
	Median 75.7	(range 66-86)	
	<75 years	8	40%
	≥75 years	12	60%
ECOG at screening			
	0	9	45%
	1	7	35%
	2	3	15%
	Unknown	1	5%
% blasts at screening (bone marrow)			
	Median 34	(range 6-94)	
	<20%	6	30%
	≥20%	13	65%
	Unknown	1	5%
No. lines previous therapies			
	Median 1	(range 1-8)	
	1	12	60%
	2	6	30%
	≥3	2	10%

Patients with relapsed AML – patients who have achieved an objective response to their most recent AML treatment

Disease Diagnosis (relapsed only)

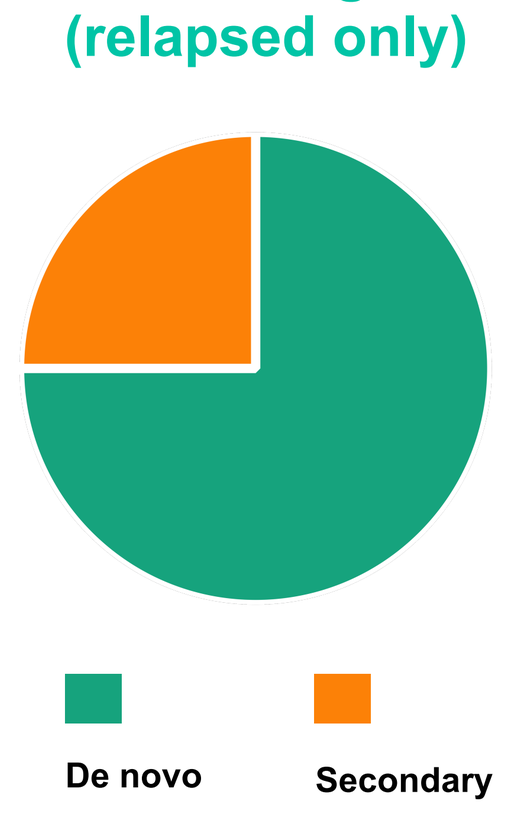

De novo
Secondary

Cytogenetic Risk Class (relapsed only)

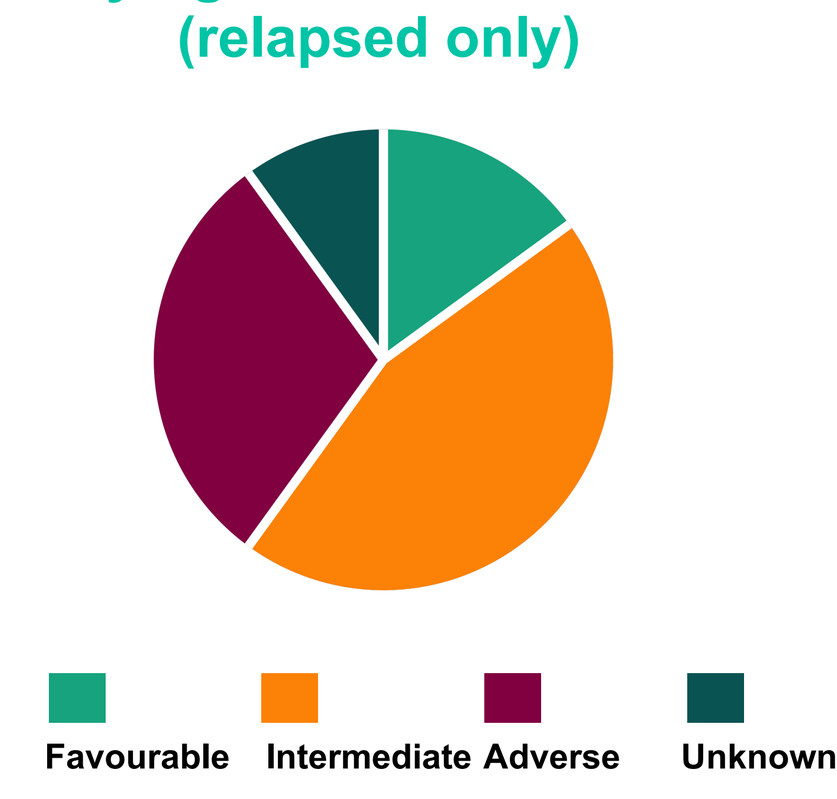

Favourable
Intermediate
Adverse
Unknown

Bemcentinib + LDAC combination is well tolerated in this elderly, frail AML population

Safety profile of combination treatment consistent with that of the individual drugs

TRAEs in ≥ 10% patients

All patients in B2 and B5 (n=34)

Preferred term	Any grade n (%)	Grades ≥3 n (%)
Hematologic		
Anaemia	7 (21)	7 (21)
Platelet count decreased/Thrombocytopenia	6 (18)	6 (18)
Non-hematologic		
Electrocardiogram QT prolonged*	16 (47)	4 (12)
Diarrhoea	8 (25)	0
Nausea	7 (21)	1 (3)
Vomiting	5 (16)	0

* No AEs of QT prolongation >grade 3 were observed

Relapsed AML pts in B2 and B5 (n=20)

Preferred term	Any grade n (%)	Grades ≥3 n (%)
Hematologic		
Anaemia	4 (20)	4 (20)
Platelet count decreased/ Thrombocytopenia	3 (15)	3 (15)
Non-hematologic		
Electrocardiogram QT prolonged*	11 (55)	3 (15)
Diarrhoea	5 (25)	0
Nausea	5 (25)	1 (5)
Vomiting	4 (20)	0
Mouth ulceration	2 (10)	1 (5)
Decreased appetite	2 (10)	0
Fatigue	2 (10)	0

Data cut-off 30th April 2021

RESULTS

Time on treatment

Parts B2+B5 (efficacy evaluable* subjects n=16 relapsed, n=7 refractory)

Response	Relapsed disease (n=16)
ORR rate (CR/CRi/PR)	5/16 (31%)
SD rate	4/16 (25%)
Clinical benefit rate (responses + SD)	9/19 (56%)

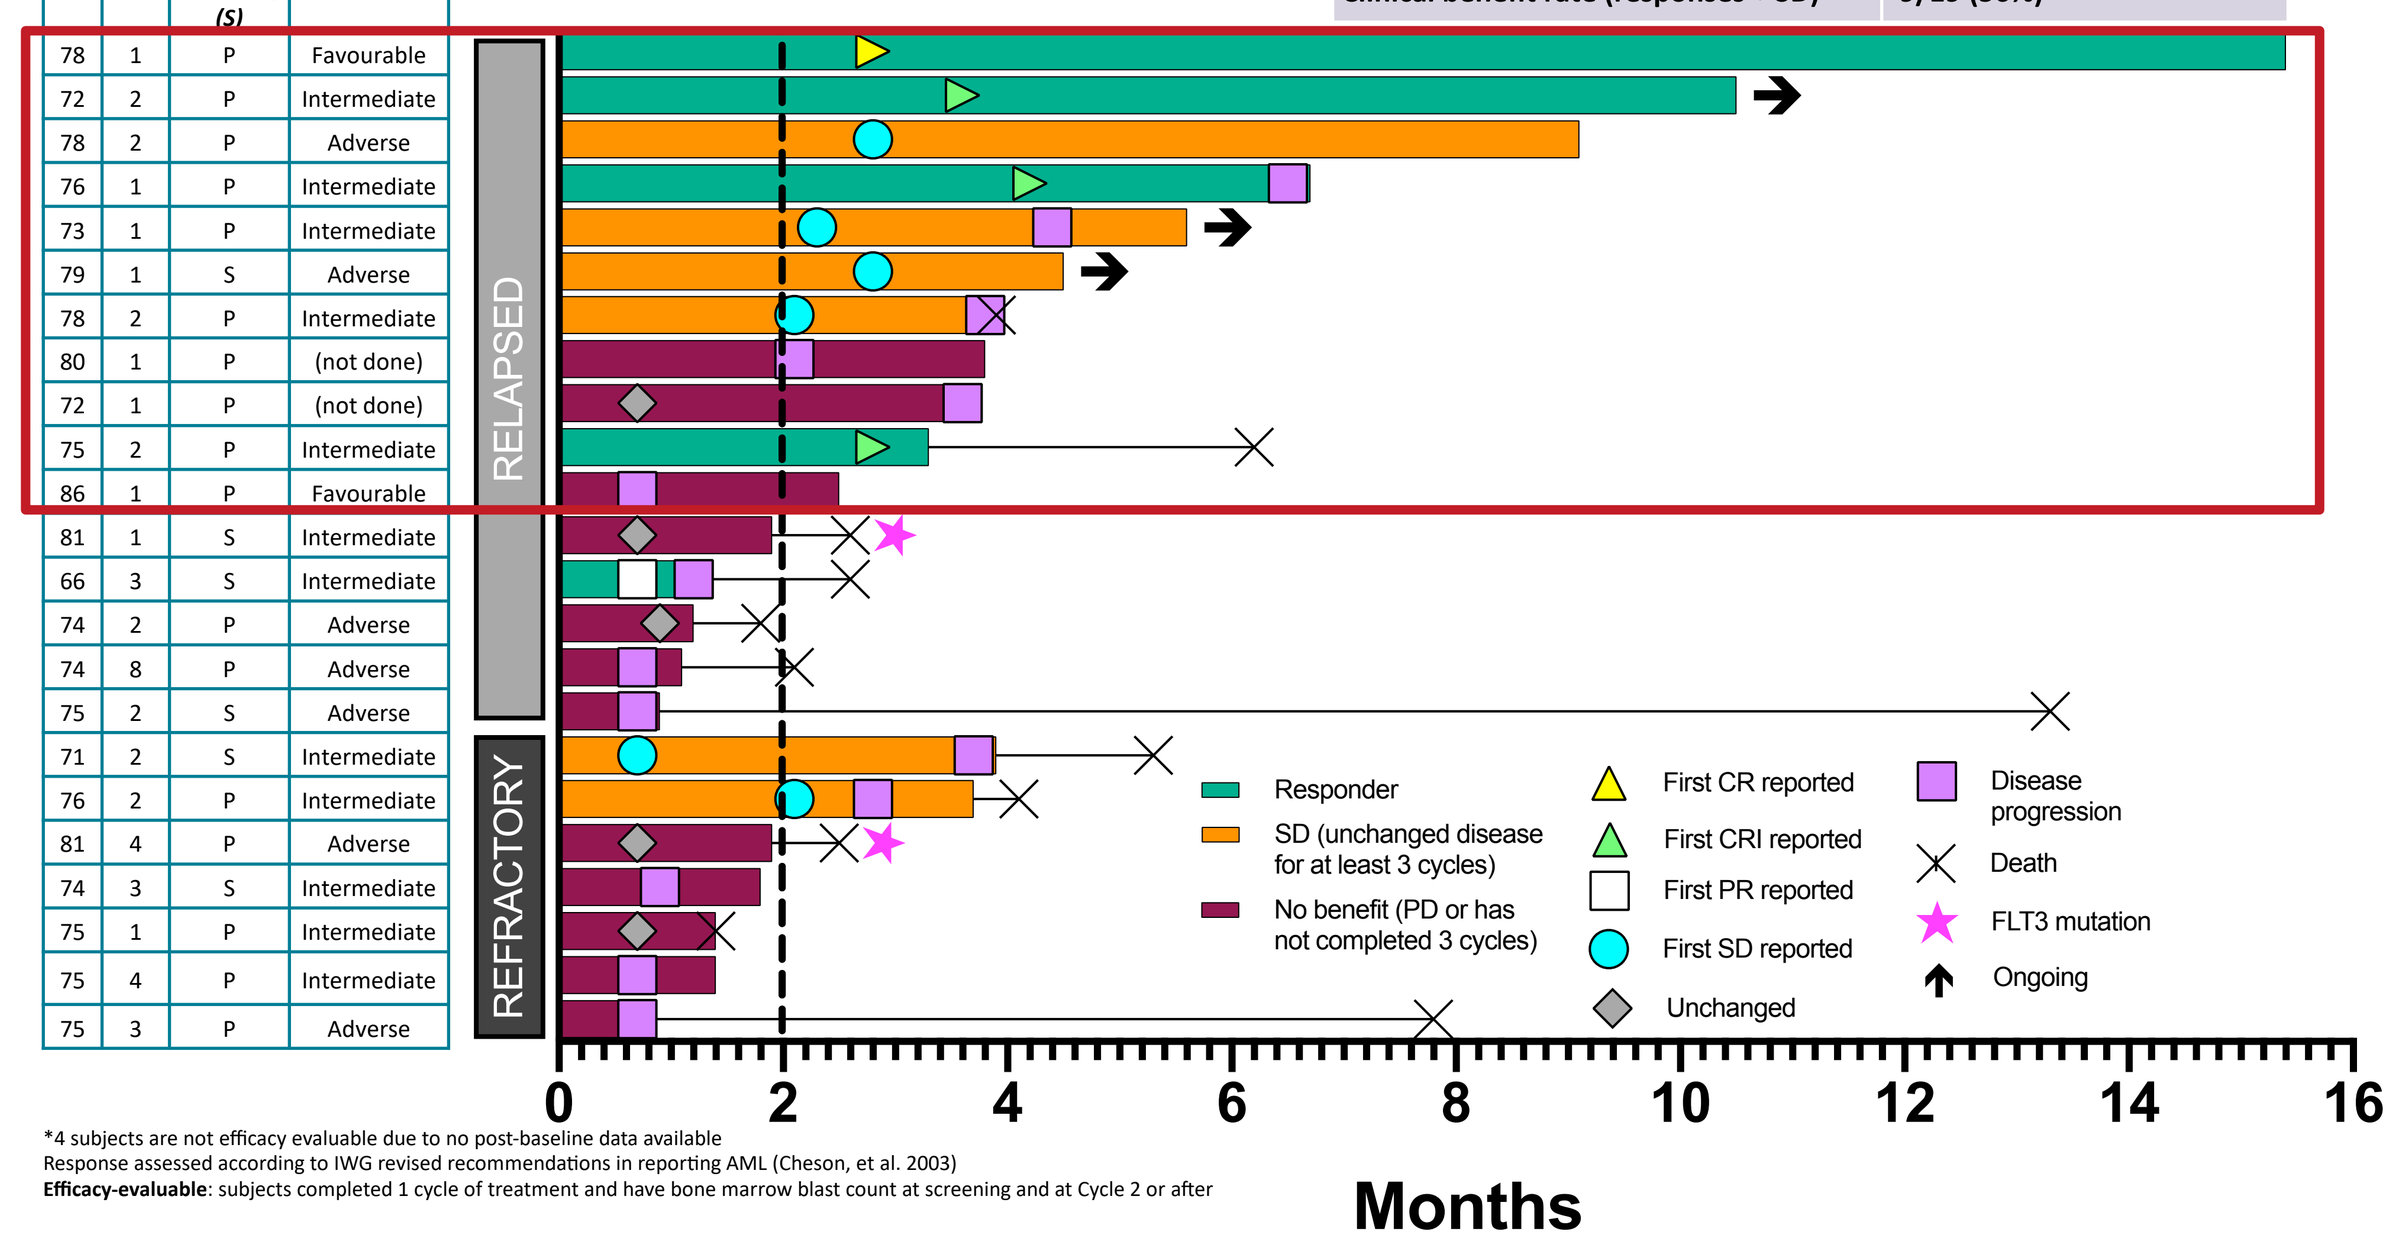

Age
Lines prior Tx
Disease type Primary (P) or Secondary (S)
Cytogenetic risk
78 1 P Favourable
72 2 P Intermediate
78 2 P Adverse
76 1 P Intermediate
73 1 P Intermediate
79 1 S Adverse
78 2 P Intermediate
80 1 P (not done)
72 1 P (not done)
75 2 P Intermediate
86 1 P Favourable
81 1 S Intermediate
66 3 S Intermediate
74 2 P Adverse
74 8 P Adverse
75 2 S Adverse
71 2 S Intermediate
76 2 P Intermediate
81 4 P Adverse
74 3 S Intermediate
75 1 P Intermediate
75 4 P Intermediate
75 3 P Adverse
RELAPSED
REFRACTORY
Responder
SD (unchanged disease for at least 3 cycles)
No benefit (PD or has not completed 3 cycles)
First CR reported
First CRI reported
First PR reported
First SD reported
Unchanged
Disease progression
Death
FLT3 mutation
Ongoing
0
2
4
6
8
10
12
14
16
Months

*4 subjects are not efficacy evaluable due to no post-baseline data available
Response assessed according to IWG revised recommendations in reporting AML (Cheson, et al. 2003)
Efficacy-evaluable: subjects completed 1 cycle of treatment and have bone marrow blast count at screening and at Cycle 2 or after

Parts B2+B5, relapsed pts with ≥ 2 cycles of Bemcentinib+LDAC n=11*

*Subset of patients who demonstrated increased clinical benefit and response rates had the following characteristics:
- Continued treatment beyond 8 weeks (56 days), and
- received ≥2 cycles of Bem+LDAC
- BM assessment beyond 8 weeks

Response	Relapsed disease (n=11)
CR/CRi rate	4/11 (36%)
SD rate	4/11 (36%)
Clinical benefit rate (responses + SD)	8/11 (73%)

Median time on treatment	Median time on treatment
8.6 months	3.2 months

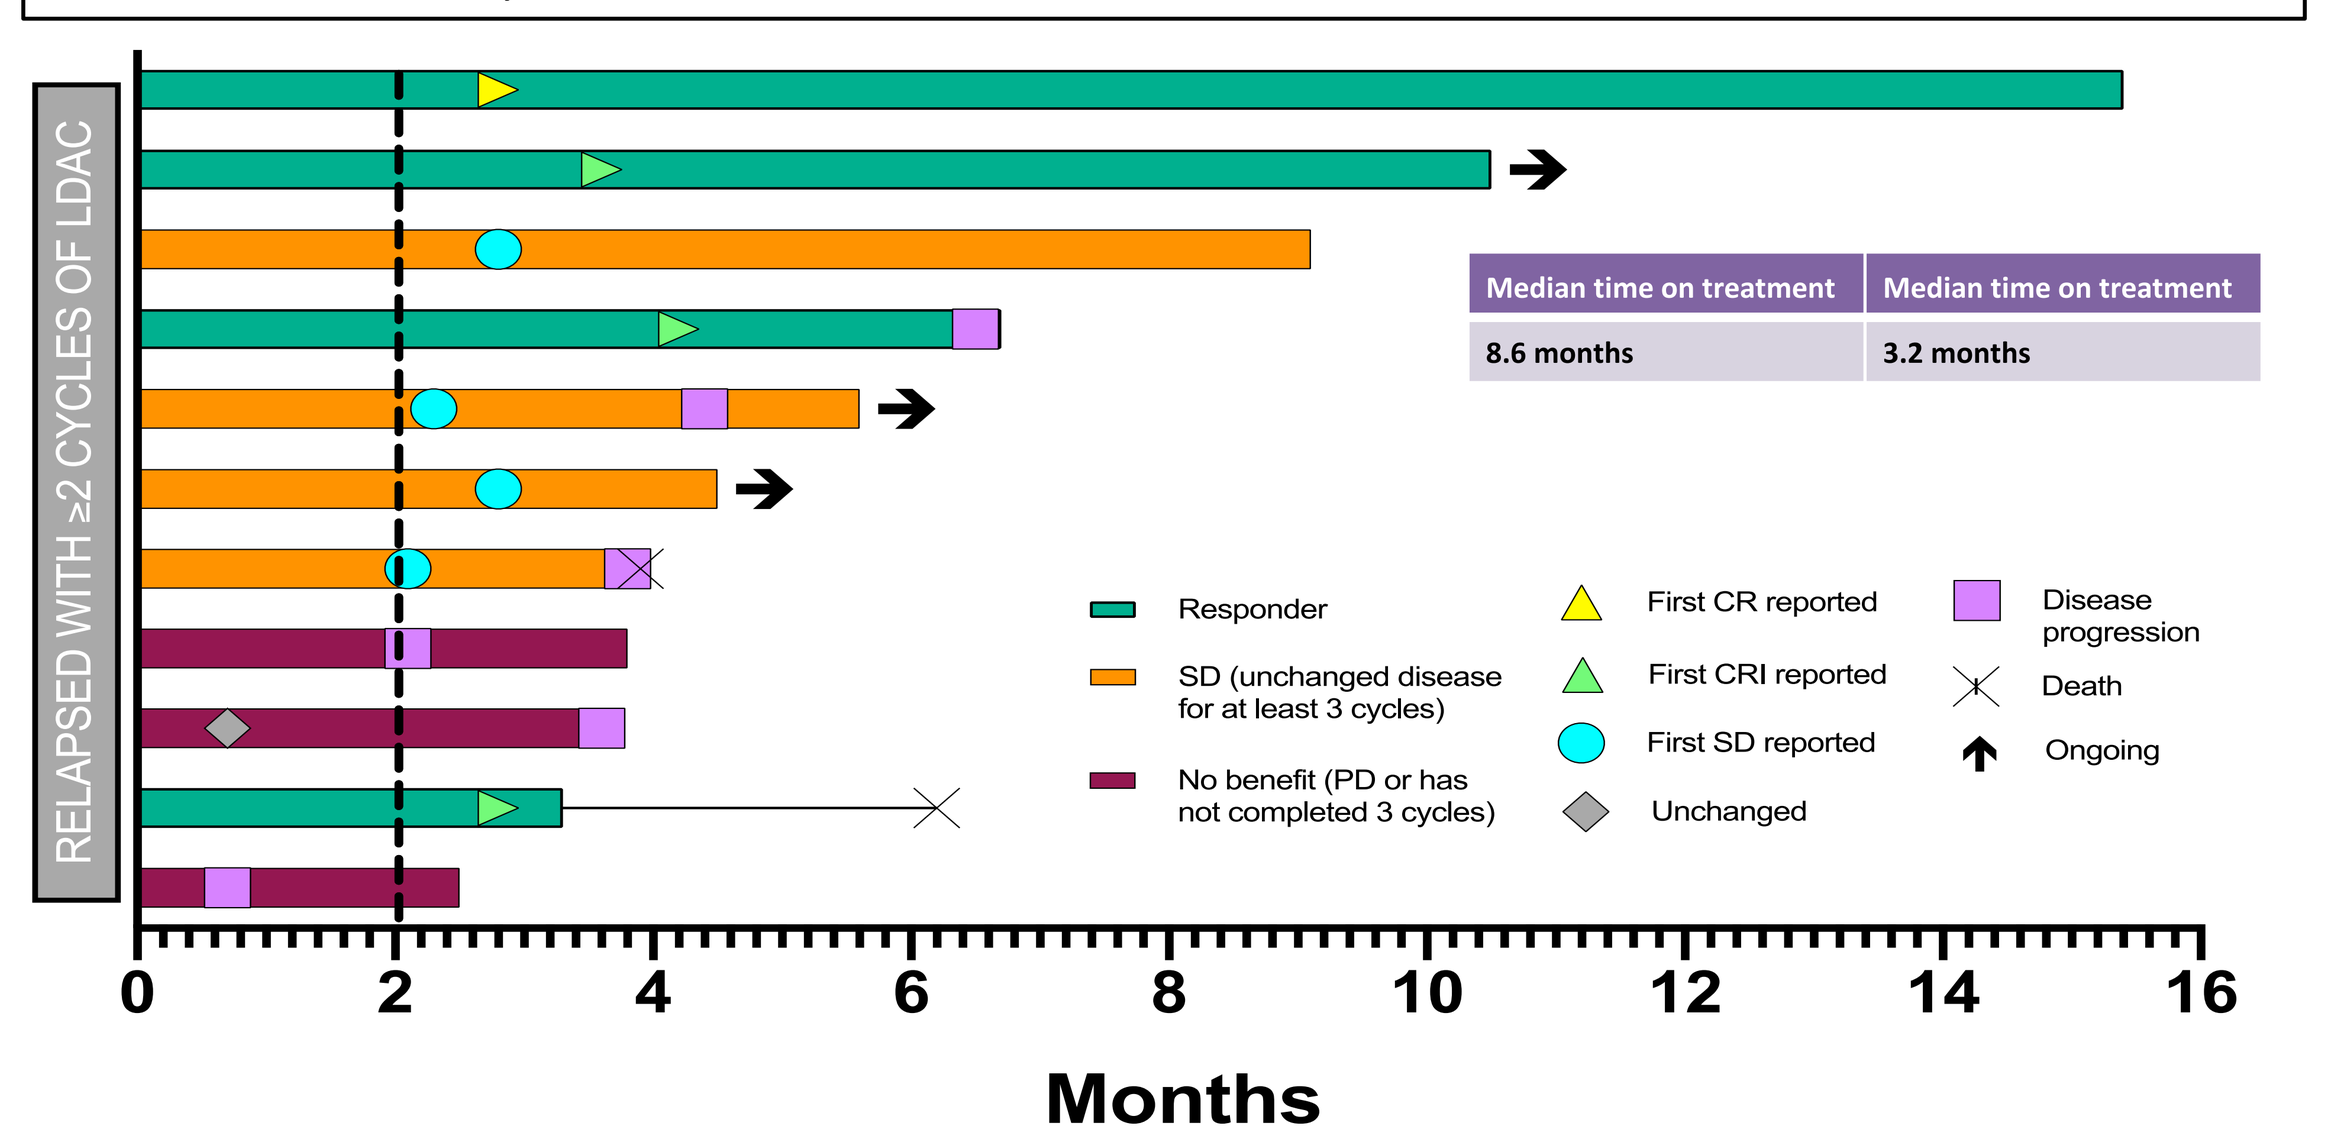

RELAPSED WITH ≥2 CYCLES OF LDAC
Responder
SD (unchanged disease for at least 3 cycles)
No benefit (PD or has not completed 3 cycles)
First CR reported
First CRI reported
First SD reported
Unchanged
Disease progression
Death
Ongoing
0
2
4
6
8
10
12
14
16
Months

Survival

Relapsed AML pts in B2 and B5 cohorts (evaluable pts n=16)

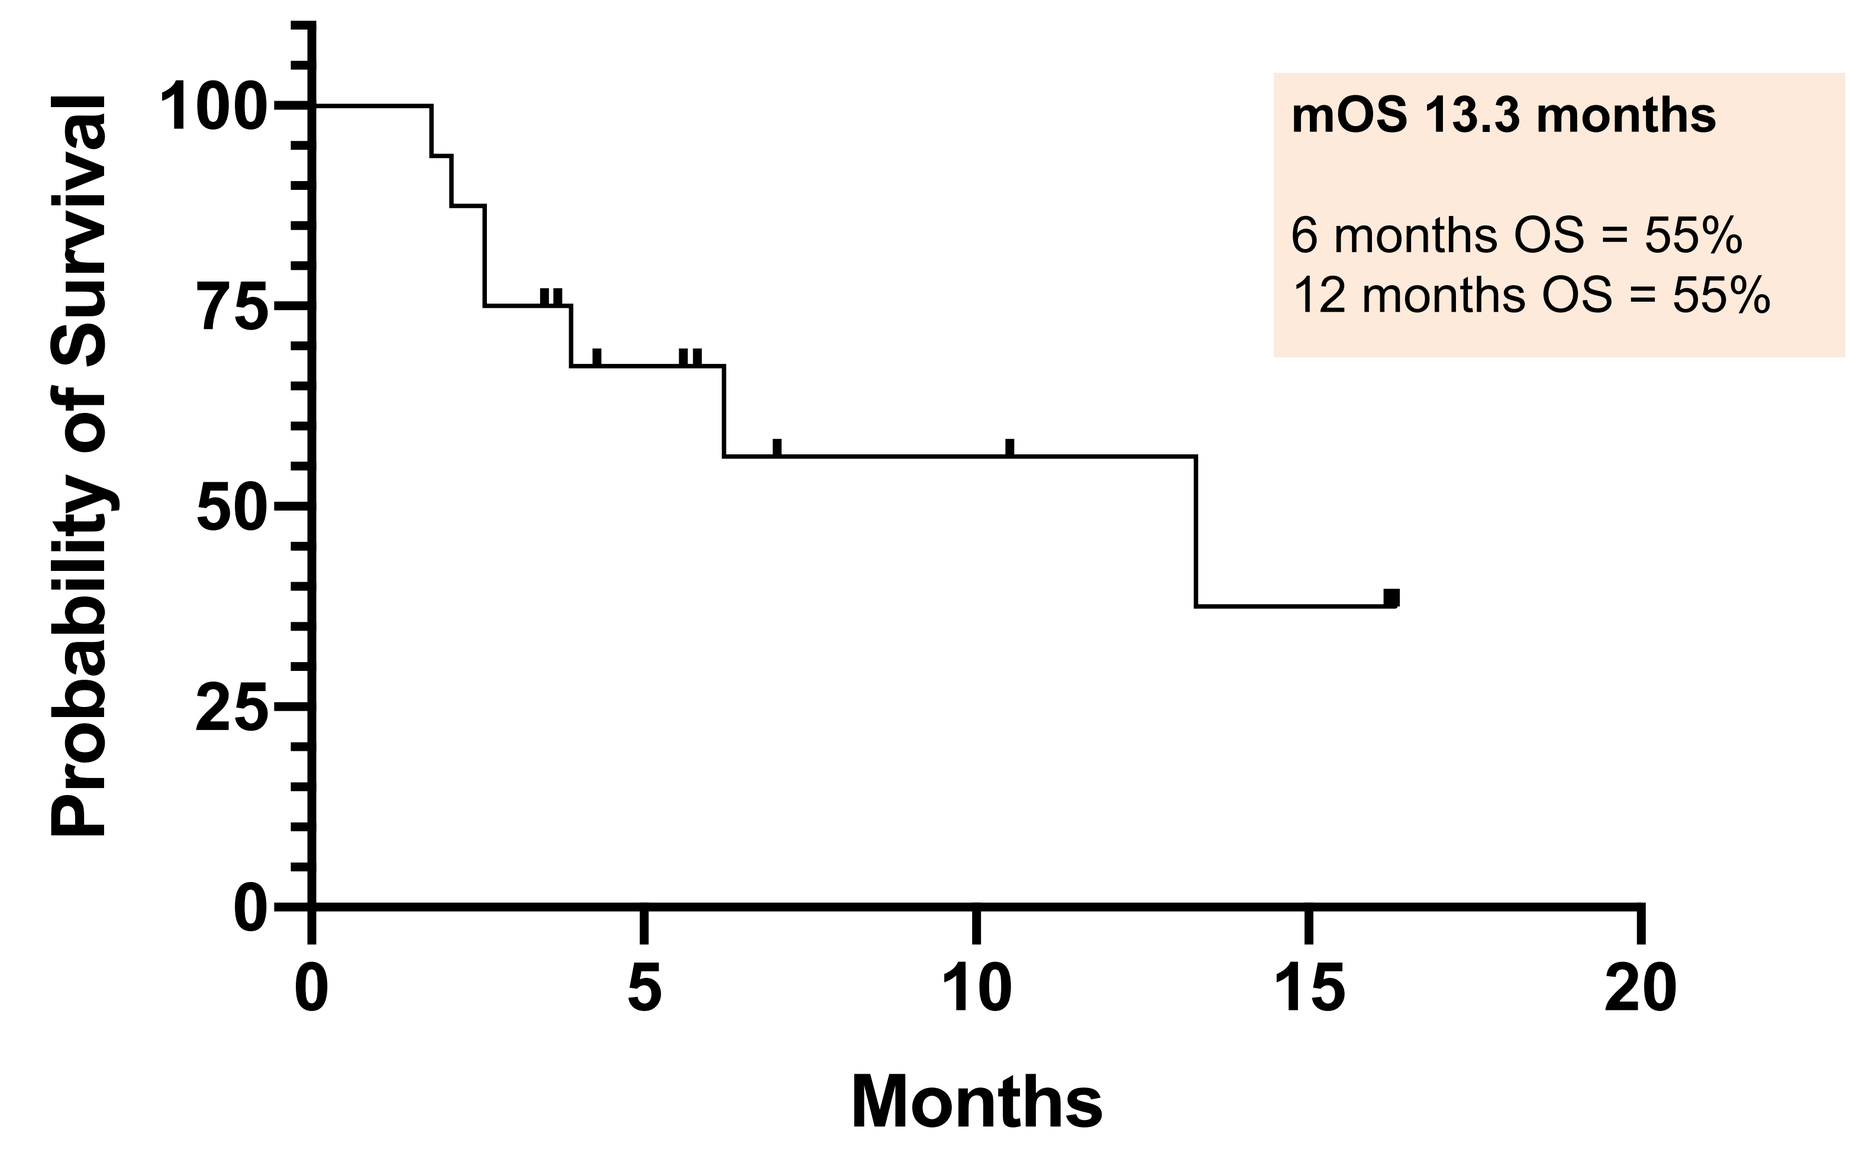

Probability of Survival
100
75
50
25
0
0
5
10
15
20
Months
mOS 13.3 months
6 months OS = 55%
12 months OS = 55%

9 pts remain alive, 3 pts ongoing on treatment

Relapsed AML pts in B2 and B5 cohorts treated with ≥ 2 cycles of Bemcentinib+LDAC (evaluable pts n=11

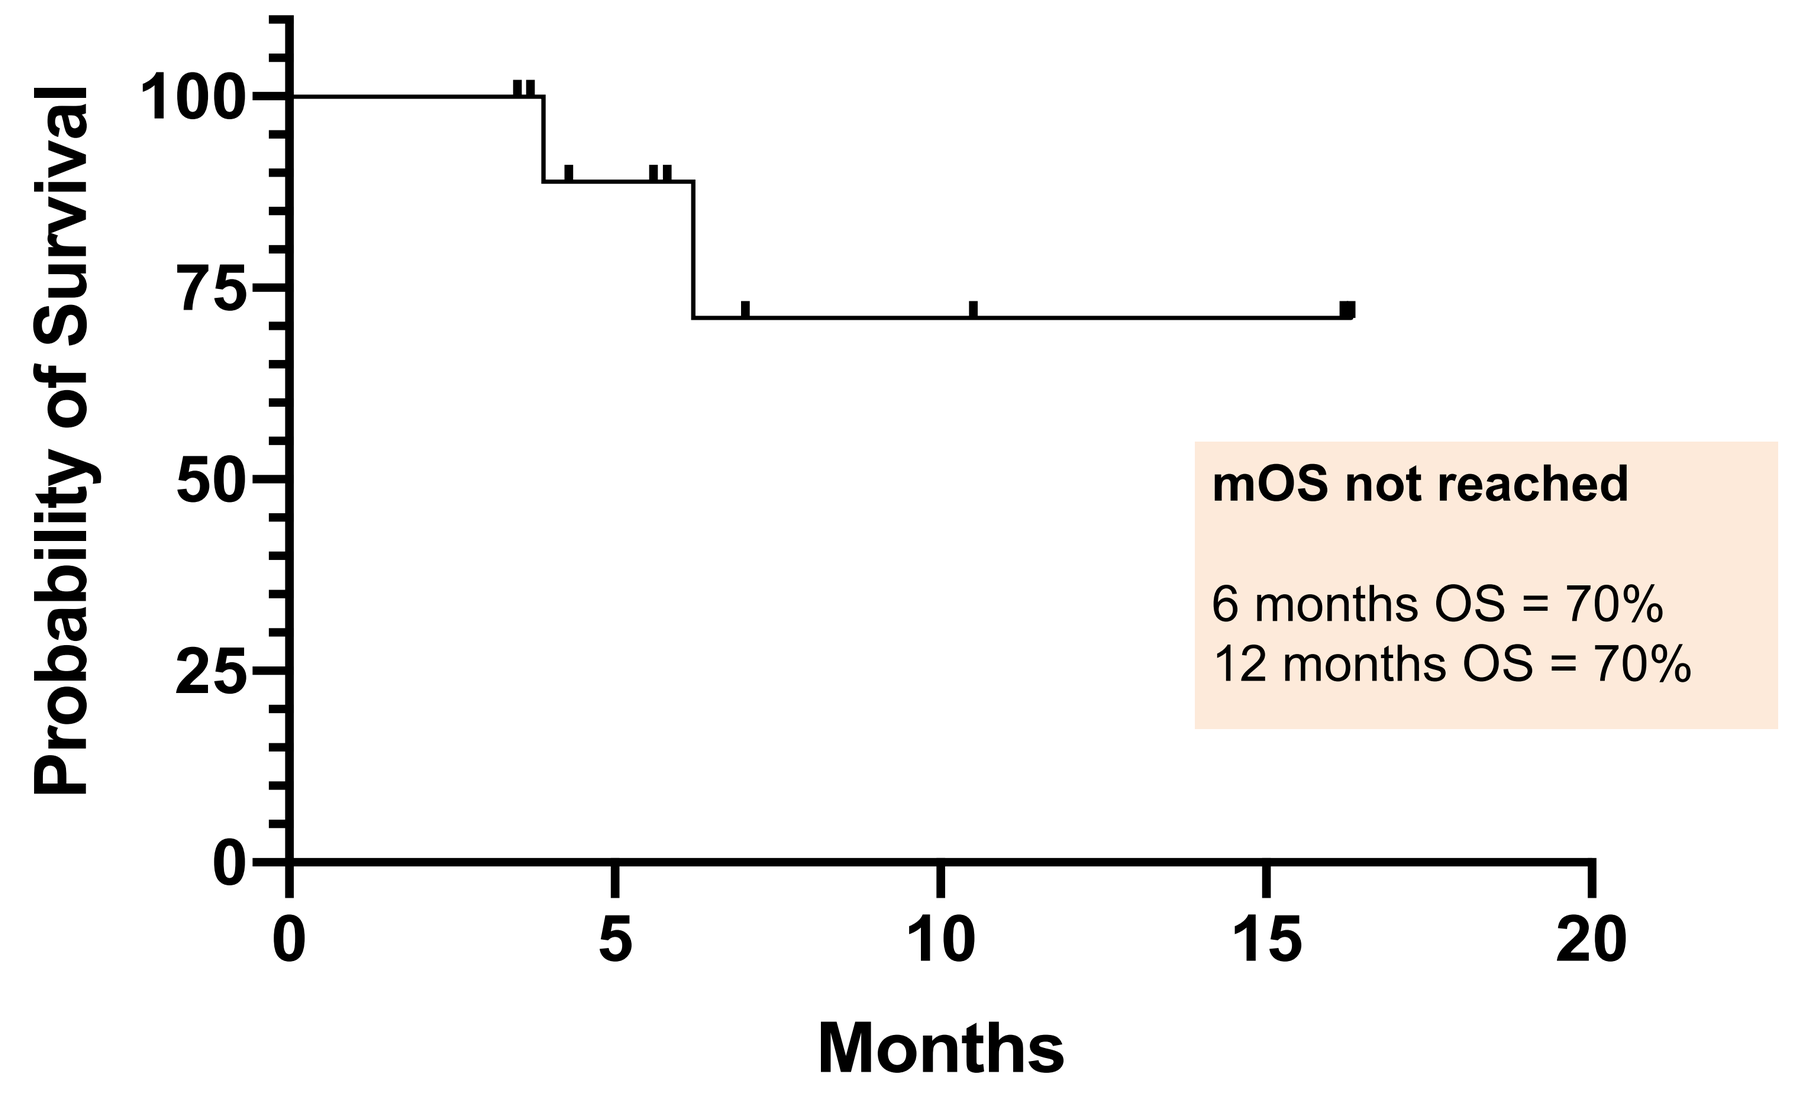

Probability of Survival
100
75
50
25
0
0
5
10
15
20
Months
mOS not reached
6 months OS = 70%
12 months OS = 70%

6 pts have discontinued treatment, 3 pts are ongoing, 9 pts remain alive

Single cell multiomics of longitudinal patient bone marrow samples

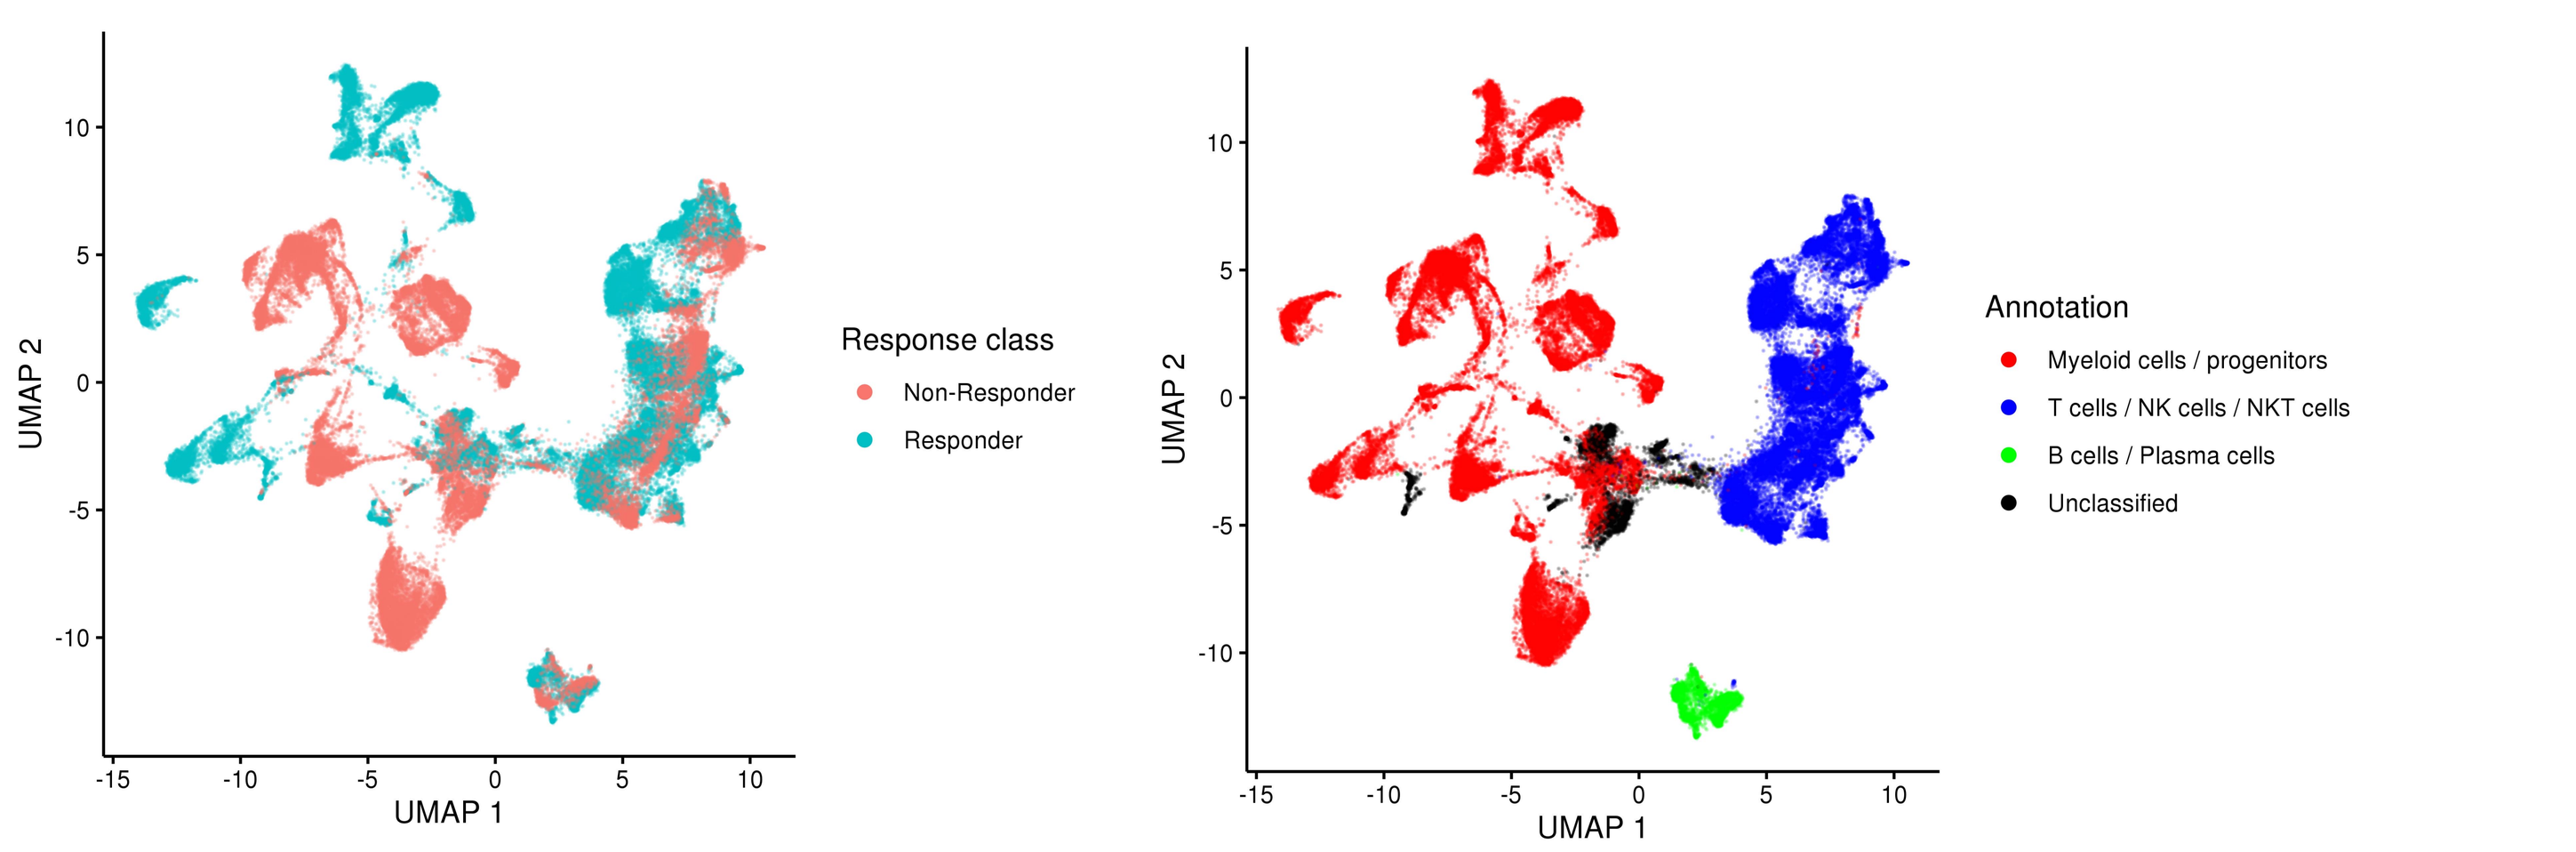

UMAP 2
UMAP 1
Response class
Non-Responder
Responder
Annotation
Myeloid cells / progenitors
T cells / NK cells / NKT cells
B cells / Plasma cells
Unclassified

Sub-clustering of the T-& NK-cell compartment

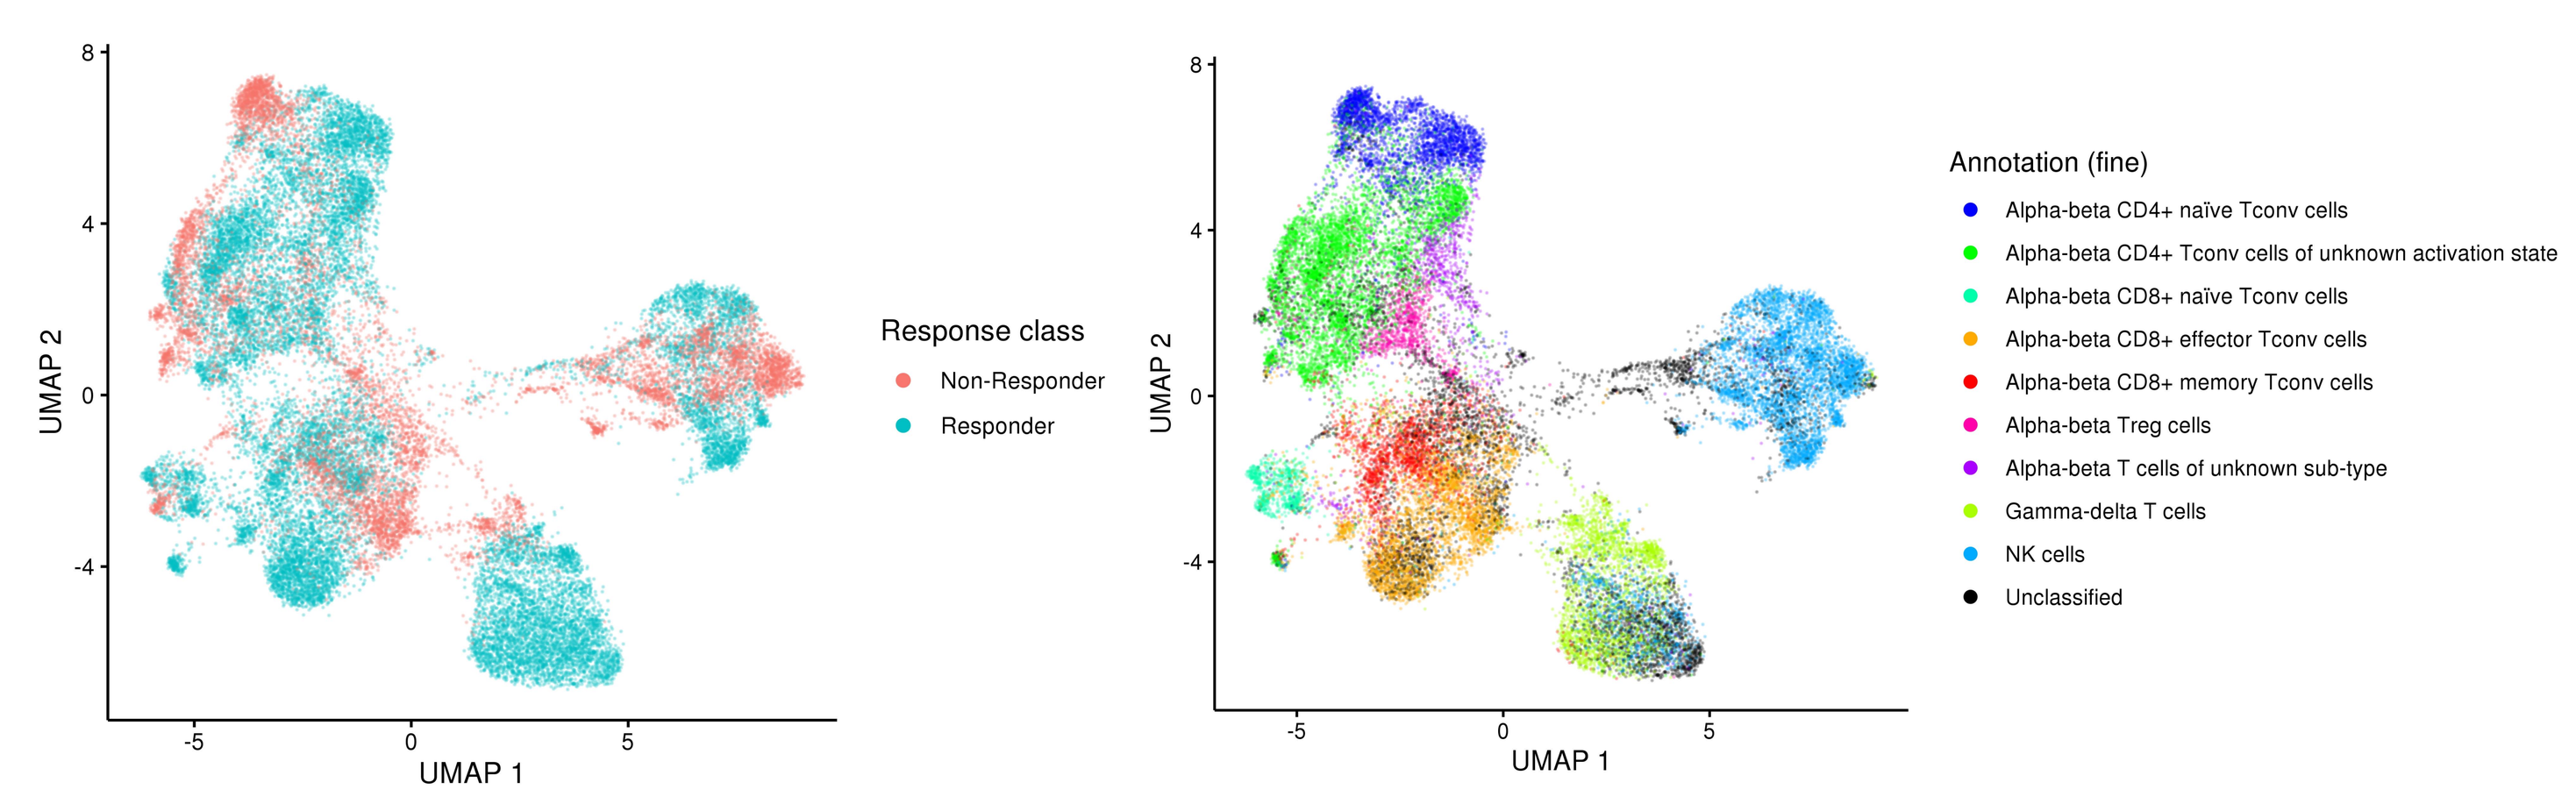

UMAP 2
UMAP 1
Response class
Non-Responder
Responder
Annotation (fine)
Alpha-beta CD4+ naïve Tconv cells
Alpha-beta CD4+ Tconv cells of unknown activation state
Alpha-beta CD8+ naïve Tconv cells
Alpha-beta CD8+ effector Tconv cells
Alpha-beta CD8+ memory Tconv cells
Alpha-beta Treg cells
Alpha-beta T cells of unknown sub-type
Gamma-delta T cells
NK cells
Unclassified

- 10x gene expression has been performed on 36 longitudinal bone marrow samples of 16 patients within the combinational arm B2 & B5.
- Proteomics were acquired using a panel of 25 TotalSeq® antibodies.
- Gene and protein expression counts were obtained from sequencing data using Cell Ranger and cell doublets were inferred using Scrublet.
- UMAP visualizations and clustering results were obtained by state-of-the-art workflows for multi-modal single-cell data using functions from Seurat and scran.
- Cell type annotations were based on the identified clusters and were inferred from the expression of known marker features on both the RNA and the protein level.
- Response was defined as CR, CRi and PR
- Sub-clustering of the T-& NK-cell compartment showed differences with respect to response

CONCLUSIONS

Bemcentinib+LDAC is safe and well tolerated in elderly AML patients

Durable responses were observed with a CR/CRi rate of 36% (4/11) in patients who received 2 or more cycles of the combination therapy with encouraging survival to date

Historic controls of LDAC monotherapy report CR/CRi rates of 13-18% with mOS of 4-6 months [6, 7, 8, 9, 10]

Single cell multiomics of bone marrow samples and sub-clustering of the T-& NK-cell compartment shows differences with respect to response in the bemcentinib+LDAC cohorts

Further clinical development of this combination is warranted

ACKNOWLEDGMENTS

The patients, their families and all the investigators on the BGBC003 study.

REFERENCES

[1] **Rochlitz, *et al.*** Axl expression is associated with adverse prognosis and with expression of Bcl-2 and CD34 in de novo acute myeloid leukemia (AML): results from a multicenter trial of the Swiss Group for Clinical Cancer Research (SAKK), Leukemia volume 13,1352–1358 (1999); [2] **Ludwig, *et al*.** Small-Molecule Inhibition of Axl Targets Tumor Immune Suppression and Enhances Chemotherapy in Pancreatic Cancer. Cancer Res. 2018 Jan 1;78(1):246-255. (2016); [3] **Byers *et al.*** A Phase I/II and pharmacokinetic study of BGB324, a selective AXL inhibitor as monotherapy and combination with erlotinib in patients with advanced Non-Small Cell Lung Cancer (NSCLC) EORTC (2016) [4]**Davidsen *et al.*** Abstract 3774: BGB324, a selective small-molecule inhibitor of receptor tyrosine kinase AXL, targets tumor immune suppression and enhances immune checkpoint inhibitor efficacy. Cancer Res 2018;78(13 Suppl):Abstract nr 3774.AACR (2018); [5]**Ben-Batalla, *et al.*** Axl, a prognostic and therapeutic target in acute myeloid leukemia mediates paracrine crosstalk of leukemia cells with bone marrow stroma. Blood. 2013 Oct 3;122(14):2443-52. (2013). [6]**Sarkozy, *et al*.** Outcome of older patients with acute myeloid leukemia in first relapse. Am J Hematol. 2013 Sep;88(9):758-64. (2013). [7]**Pemmaraju, *et al.*** Improving outcomes for patients with acute myeloid leukemia in first relapse: a single center experience. Am J Hematol. 2015 Jan;90(1):27-30. (2015). [8]**Burnett, *et al*.** A comparison of low-dose cytarabine and hydroxyurea with or without all-trans retinoic acid for acute myeloid leukemia and high-risk myelodysplastic syndrome in patients not considered fit for intensive treatment. (2007). [9]**Stone, *et al*.** Efficacy outcomes in the treatment of older or medically unfit patients with acute myeloid leukaemia: A systematic review and meta-analysis. Leuk Res. 2019 Jul;82:36-42. (2019). [10]**Wei *et al.*** Venetoclax plus LDAC for newly diagnosed AML ineligible for intensive chemotherapy: a phase 3 randomized placebo-controlled trial. Blood. 2020 Jun 11;135(24):2137-2145.

CONTACT INFORMATION

BerGenBio Ltd.
1 Robert Robinson Ave
Oxford
OX4 4GA UK
post@bergenbio.com
+47 559 61 159

BerGenBio ASA
Jonas Lies vei 91 5009
Bergen
Norway
www.bergenbio.com
@BGenBio

Sonja Loges
University Medical Center Mannheim
68167 Mannheim, Germany
sonia.loges@medma.uni-Heidelberg.de
+49 1762 20 43 855

Disclosure
Prof Loges receives research funding, advisory board honoraria and travel support from BerGenBio.

| Connecting Hematology For Clinical and Research Excellence

EHA2021 VIRTUAL JUNE 9 - 17, 2021