Durable Responses Observed in Elderly AML Patients Unfit for Intensive Chemotherapy with First-in-Class Selective AXL Inhibitor Bemcentinib (BGB324) in Combination with LDAC: Phase II Open-Label Study

Abstract 3943

Sonja Loges[1], Michael Heuser[2], Jörg Chromik[3], Carlos Enrique Vigil[4], Peter Paschka[5], Francesca Re[6], Nicola Di Renzo[7], Roberto Lemoli[8], Daniele Mattei[9], Isabel Ben Batalla[1], Monica Hellesøy[10], David Micklem[11], Robert J Holt[11], James B. Lorens[12], Muhammad Shoaib[11], Hassan Aly[11], Wouter Hanekom[11], Walter M. Fiedler[1], Jorge E. Cortes[13], Bjørn T. Gjertsen[12]

[1]University Medical Center Hamburg-Eppendorf, Hamburg; [2]Hannover Medical School, Hannover, Germany; [3]University Hospital Frankfurt, Frankfurt, Germany; [4]Vanderbilt-Ingram Cancer Center, Nashville, United States; [5]University Hospital of Ulm, Ulm, Germany; [6]University of Parma, Parma, Italy; [7]Vito Fazzi Hospital, Lecce, Italy; [8]University of Genoa, Genoa, Italy; [9]ASO S. Croce e Carle, Cuneo, Italy; [10]Haukelands University Hospital, Bergen, Norway; [11]BerGenBio ASA, Bergen, Norway; [12]University of Bergen, Bergen, Norway; [13]The University of Texas MD Anderson Cancer Center, Houston, United States.

Background & objective

NCT02488408: a Phase II clinical trial of selective AXL inhibitor bemcentinib and low dose cytarabine (LDAC)

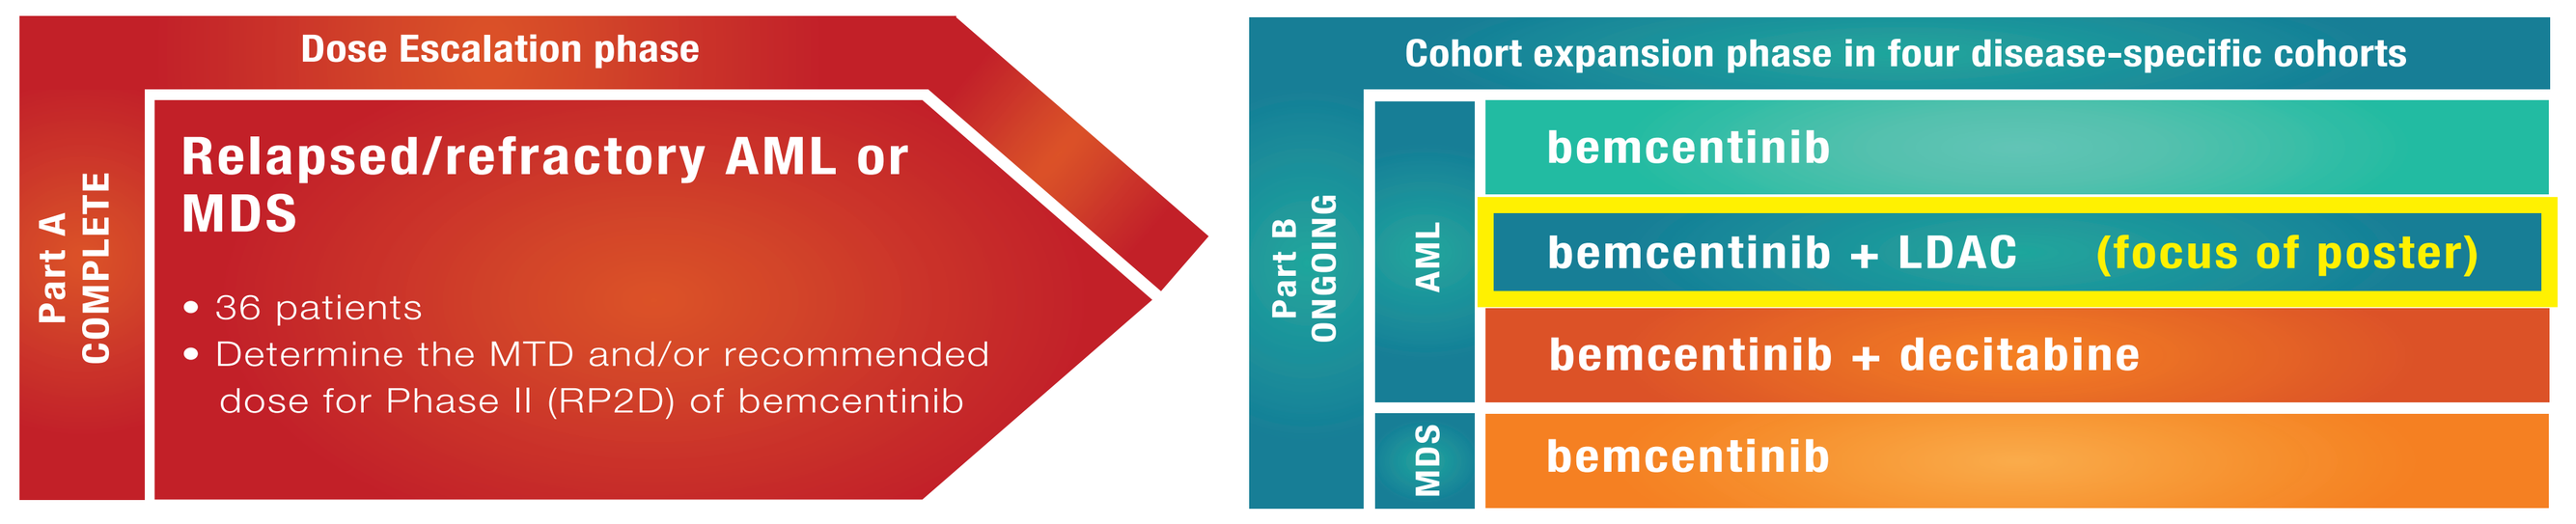
Dose Escalation phase
Part A COMPLETE
Relapsed/refractory AML or MDS
• 36 patients
• Determine the MTD and/or recommended dose for Phase II (RP2D) of bemcentinib
Cohort expansion phase in four disease-specific cohorts
Part B ONGOING
AML
MDS
bemcentinib
bemcentinib + LDAC (focus of poster)
bemcentinib + decitabine
bemcentinib

Assessments - efficacy & safety

- Response assessed according to IWG revised recommendations in reporting AML[1]
- Adverse events were assessed by CTCAE v4.0
- Results reported as of data cutoff (30 Sep. 2019)
- Safety-evaluable: >/= dose of study treatment
- Efficacy-evaluable: subjects completed 1 cycle of treatment and have bone marrow blast count at screening and at Cycle 2 or after

Biomarker analysis

- Soluble protein biomarker analsis from plasma samples

Endpoints

- **Primary:** safety and tolerability
- **Secondary:** ORR, RFS, OS, PK profile

Key inclusion criteria

- Histological, molecular, or cytological confirmation of AML (with the exception of AML M3)
- Patients with AML who are not suitable for intensive chemotherapy as a result of advanced age or co-morbidities and who are suitable to receive treatment with cytarabine
- Eastern Cooperative Oncology Group (ECOG) performance status 0,1 or 2

Key exclusion criteria

- Patients with a matched donor who are candidates for allogenic BM transplantation
- Abnormal left ventricular ejection fraction on echocardiography (less than the lower limit of normal for a patient of that age at the treating institution or <45%, whichever is lower)
- Current treatment with any agent known to cause Torsades de pointes which cannot be discontinued at least five half-lives or two weeks prior to first dose of study treatment
- Impaired renal function as demonstrated by a creatinine clearance of <30 mL/min determined by Cockcroft-Gault formula
- Radiotherapy or chemotherapy within the 14 days prior to the first dose of bemcentinib being administered (other than hydroxyurea)
- Receiving investigational anti-cancer treatment concurrently or within 14 days or five half-lives of either the parent drug or any active metabolite prior to starting bemcentinib
- Unresolved CTCAE ≥Grade 2 toxicity (other than stable toxicity) from previous anti-cancer therapy excluding alopecia
- Prior exposure to Gilteritinib

AXL receptor tyrosine kinase, AML, and selective AXL inhibitor bemcentinib

- AXL is a receptor tyrosine kinase expressed on both tumor and immune cells that is associated with poor outcome in many cancers[2,3,4,5]
- AXL is expressed in ca. 50% of all AML cases and is a negative prognostic factor in AML[6]
- AXL is overexpressed in response to a hostile tumour microenvironment and drives a tumour survival programme resulting in immune evasion, therapy resistance & metastasis[3]
- The AXL kinase inhibitor bemcentinib is a first-in-class highly selective, potent, and orally bioavailable inhibitor of AXL.
- Bemcentinib inhibits survival of AML cells *in vitro* and enhances sensitivity toward cytarabine in fresh AML blasts and AML cell lines[7]
- Bemcentinib potentiates the efficacy of chemotherapy[8], prevents development of acquired drug resistance[9] and enhances the efficacy of immune checkpoint inhibitors[10] in several *in vivo* tumor models.
- Bemcentinib is being developed as a monotherapy and in combination with chemo-, immune-, and targeted therapy in AML/MDS, NSCLC and melanoma.

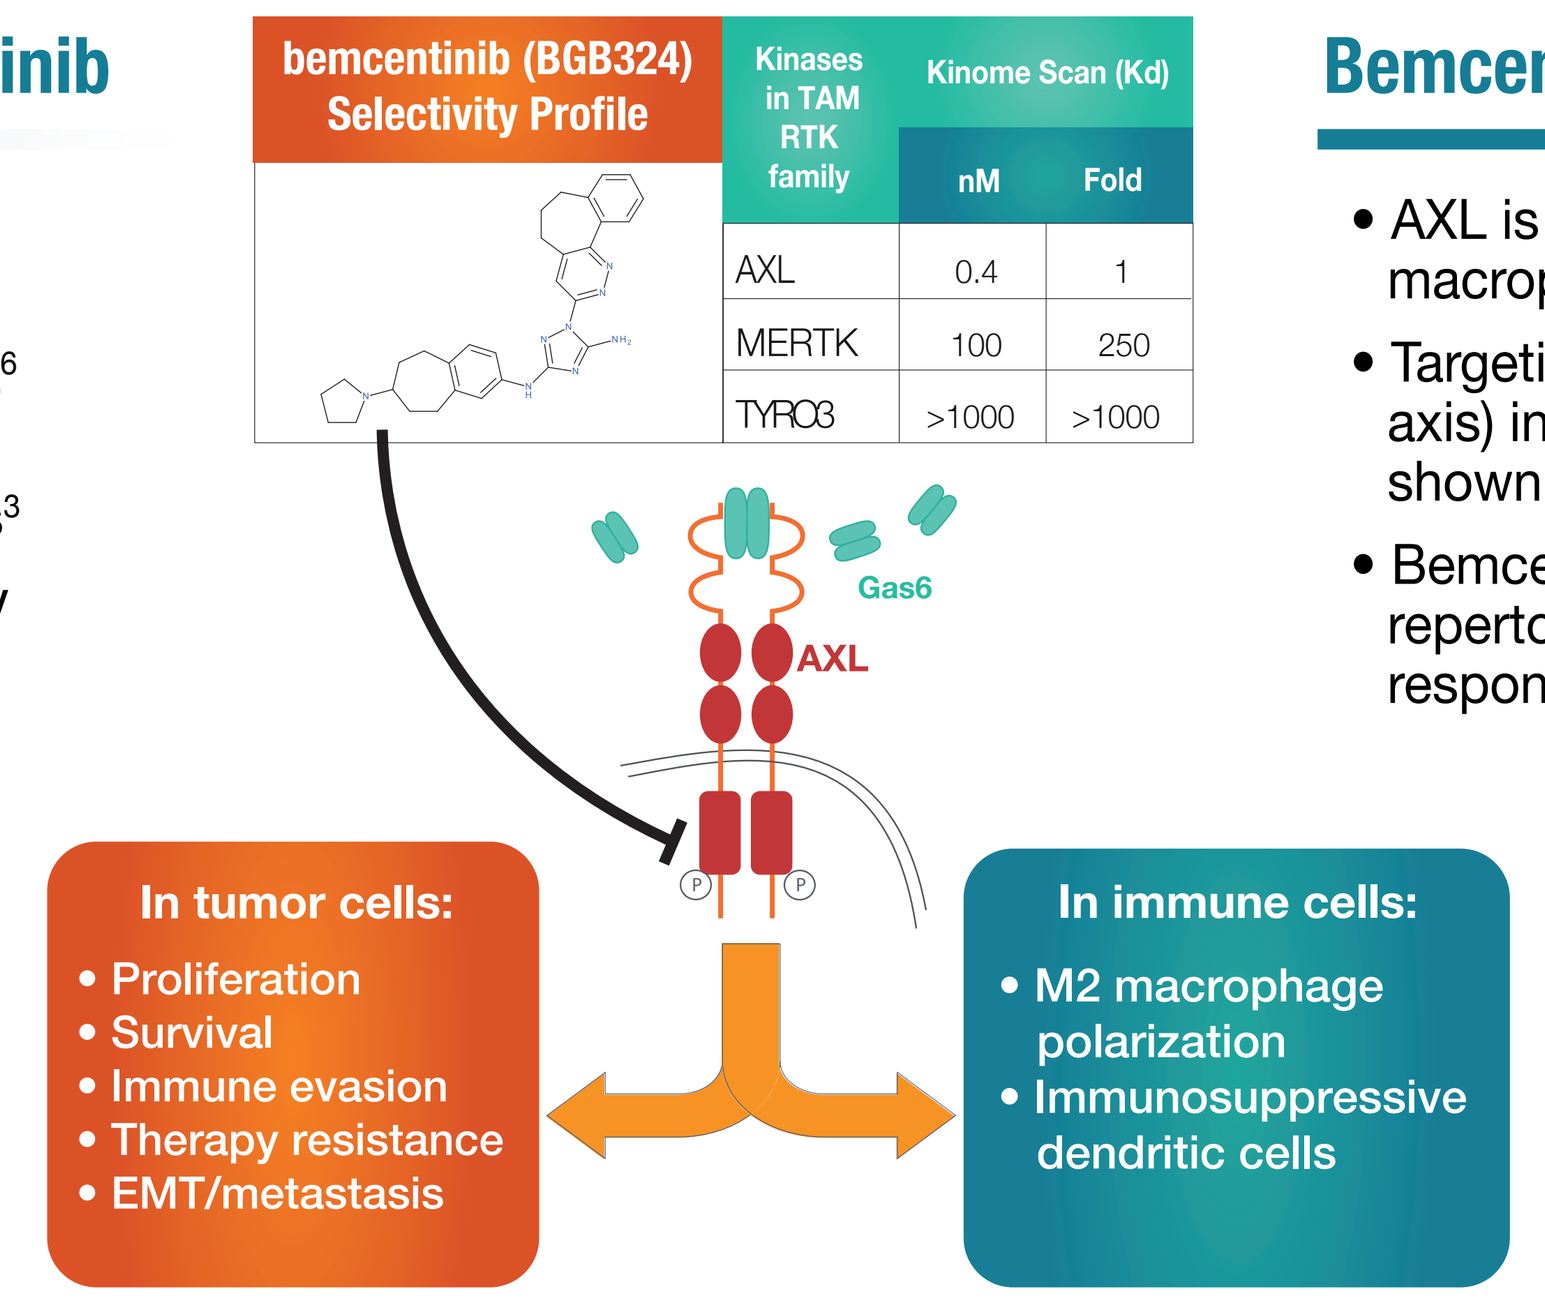
bemcentinib (BGB324) Selectivity Profile
Kinases in TAM RTK family
Kinome Scan (Kd)
nM
Fold
AXL 0.4 1
MERTK 100 250
TYRO3 >1000 >1000
Gas6
AXL
In tumor cells:
• Proliferation
• Survival
• Immune evasion
• Therapy resistance
• EMT/metastasis
In immune cells:
• M2 macrophage polarization
• Immunosuppressive dendritic cells

Bemcentinib promotes anti-tumor immunity

- AXL is expressed by suppressive tumor-associated M2 macrophages and dendritic cells in solid tumors[8]
- Targeting immunosuppressive pathways (such as PD-1/PD-L1 axis) in combination with chemotherapy in AML patients has shown promising efficacy[11]
- Bemcentinib monotherapy treatment led to T- and B-cell repertoire (TCR/BCR) diversification indicative of an immune response in 7/11 patients[12]

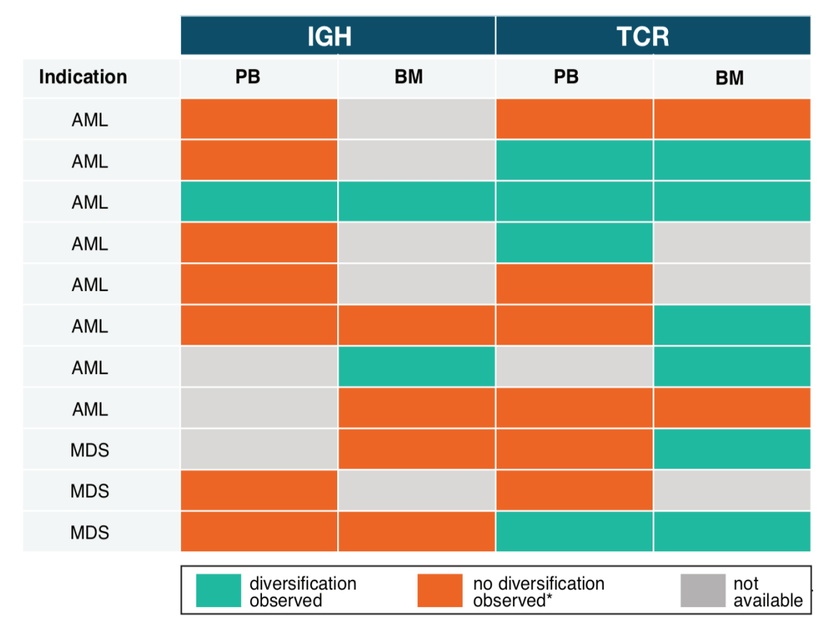
IGH
TCR
Indication
PB
BM
AML
MDS
diversification observed
no diversification observed*
not available

Results

Patient demographics and Safety

LDAC + bemcentinib (n=14*)

Characteristics	n	%
Sex		
Male	10	71%
Female	4	29%
Age		
Mean	75.5	
Median	76	
Range	66-83	
<75	4	29%
>75	10	71%
ECOG at screening		
0	3	21%
1	10	71%
2	1	7%
% blasts at screening (bone marrow)		
Median	34	
Range	3-96	
<20%	1	7%
>20%	13	93%
Disease diagnosis		
De novo	8	57%
Secondary	6	43%
FLT3 status		
Positive	1	7%
Negative	10	71%
Indeterminate/not tested	3	21%
Cytogenetic risk class		
Favourable	1	7%
Intermediate	8	57%
Poor	5	36%
No. lines previous therapy		
Median	1	
Range	0-8	
0	6	43%
1	2	14%
2	2	14%
>3	4	29%
Disease status		
Refractory	4	29%
Relapsed	4	29%
First-line	6	43%

Adverse Events	
Any Grade (in ≥20% of patients)	
Any event, n (%)	14 (100%)
Haematologic	
Anaemia	7 (50%)
Neutropenia*	4 (29%)
Platelet count decreased	4 (29%)
Thrombocytopenia	5 (36%)
Non-haematologic	
Constipation	3 (21%)
Diarrhoea	7 (44%)
Dyspnoea	3 (21%)
Electrocardiogram QT prolonged	3 (21%)
Epistaxis	3 (21%)
Fatigue	3 (21%)
Gastrointestinal haemorrhage	3 (21%)
Headache	3 (21%)
Hypokalaemia	3 (21%)
Mouth haemorrhage	3 (21%)
Nausea	4 (29%)
Oedema peripheral	4 (29%)
Pyrexia	4 (29%)
Weight Decreased	3 (21%)
Grades ≥3 (in ≥20% of patients**	
Any event, n (%)	14 (100%)
Haematologic	
Anaemia	6 (43%)
Neutropenia*	4 (29%)
Platelet count decreased	4 (29%)
Thrombocytopenia	5 (36%)
Non-haematologic	
Electrocardiogram QT prolonged	2 (14%)

***No Grade 4-5 AE's occurred in any patient*

* 14 evaluable patients (out of a total of 16 en-rolled). Evaluable patients are defined as those who have completed at least 1 cycle of treatment, and have at least 1 response assessment completed based on bone marrow aspiration results.

Clinical Activity in Newly-Diagnosed Patients

Time on treatment

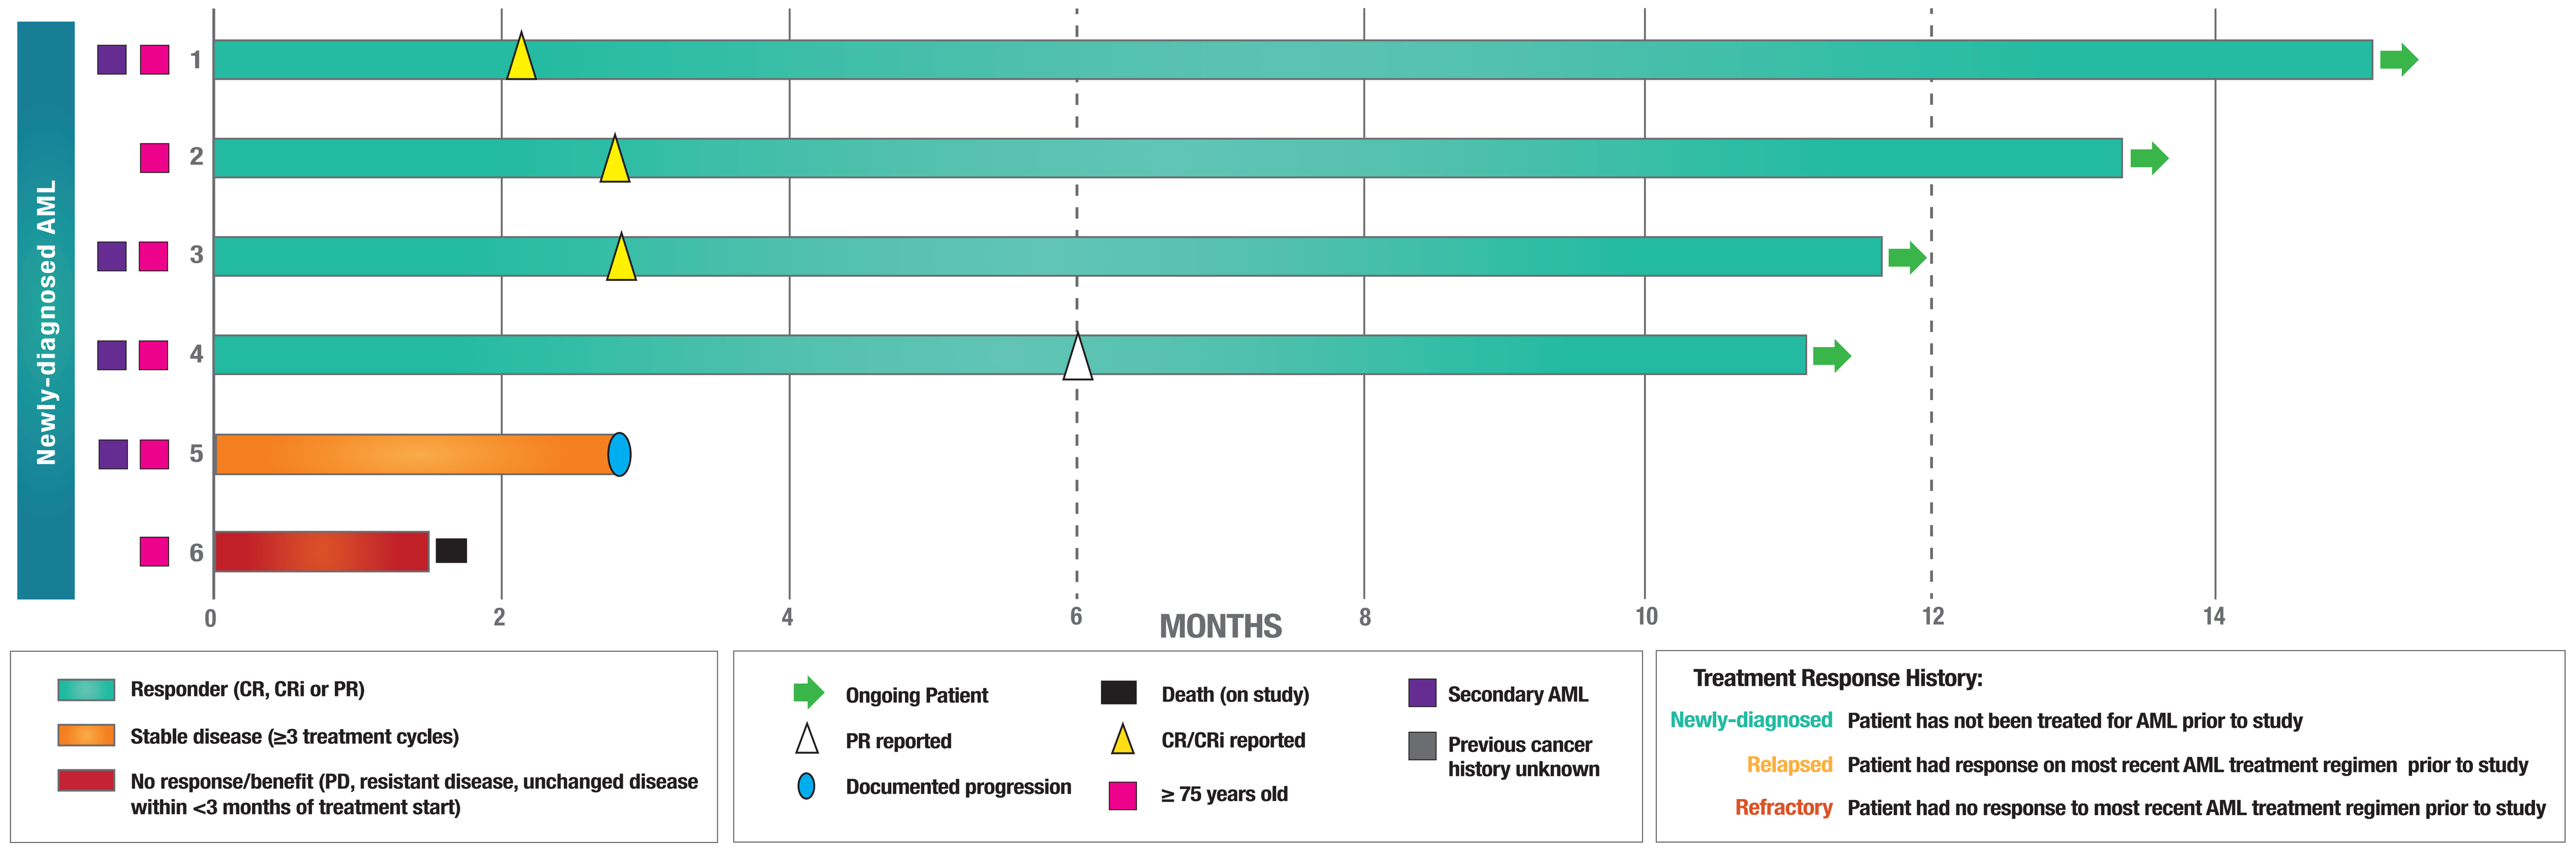
Newly-diagnosed AML
1
2
3
4
5
6
0
2
4
6
8
10
12
14
MONTHS
Responder (CR, CRi or PR)
Stable disease (≥3 treatment cycles)
No response/benefit (PD, resistant disease, unchanged disease within <3 months of treatment start)
Ongoing Patient
PR reported
Documented progression
Death (on study)
CR/CRi reported
≥ 75 years old
Secondary AML
Previous cancer history unknown
Treatment Response History:
Newly-diagnosed Patient has not been treated for AML prior to study
Relapsed Patient had response on most recent AML treatment regimen prior to study
Refractory Patient had no response to most recent AML treatment regimen prior to study

Clinical Activity in Relapsed/Refractory Patients

Time on treatment

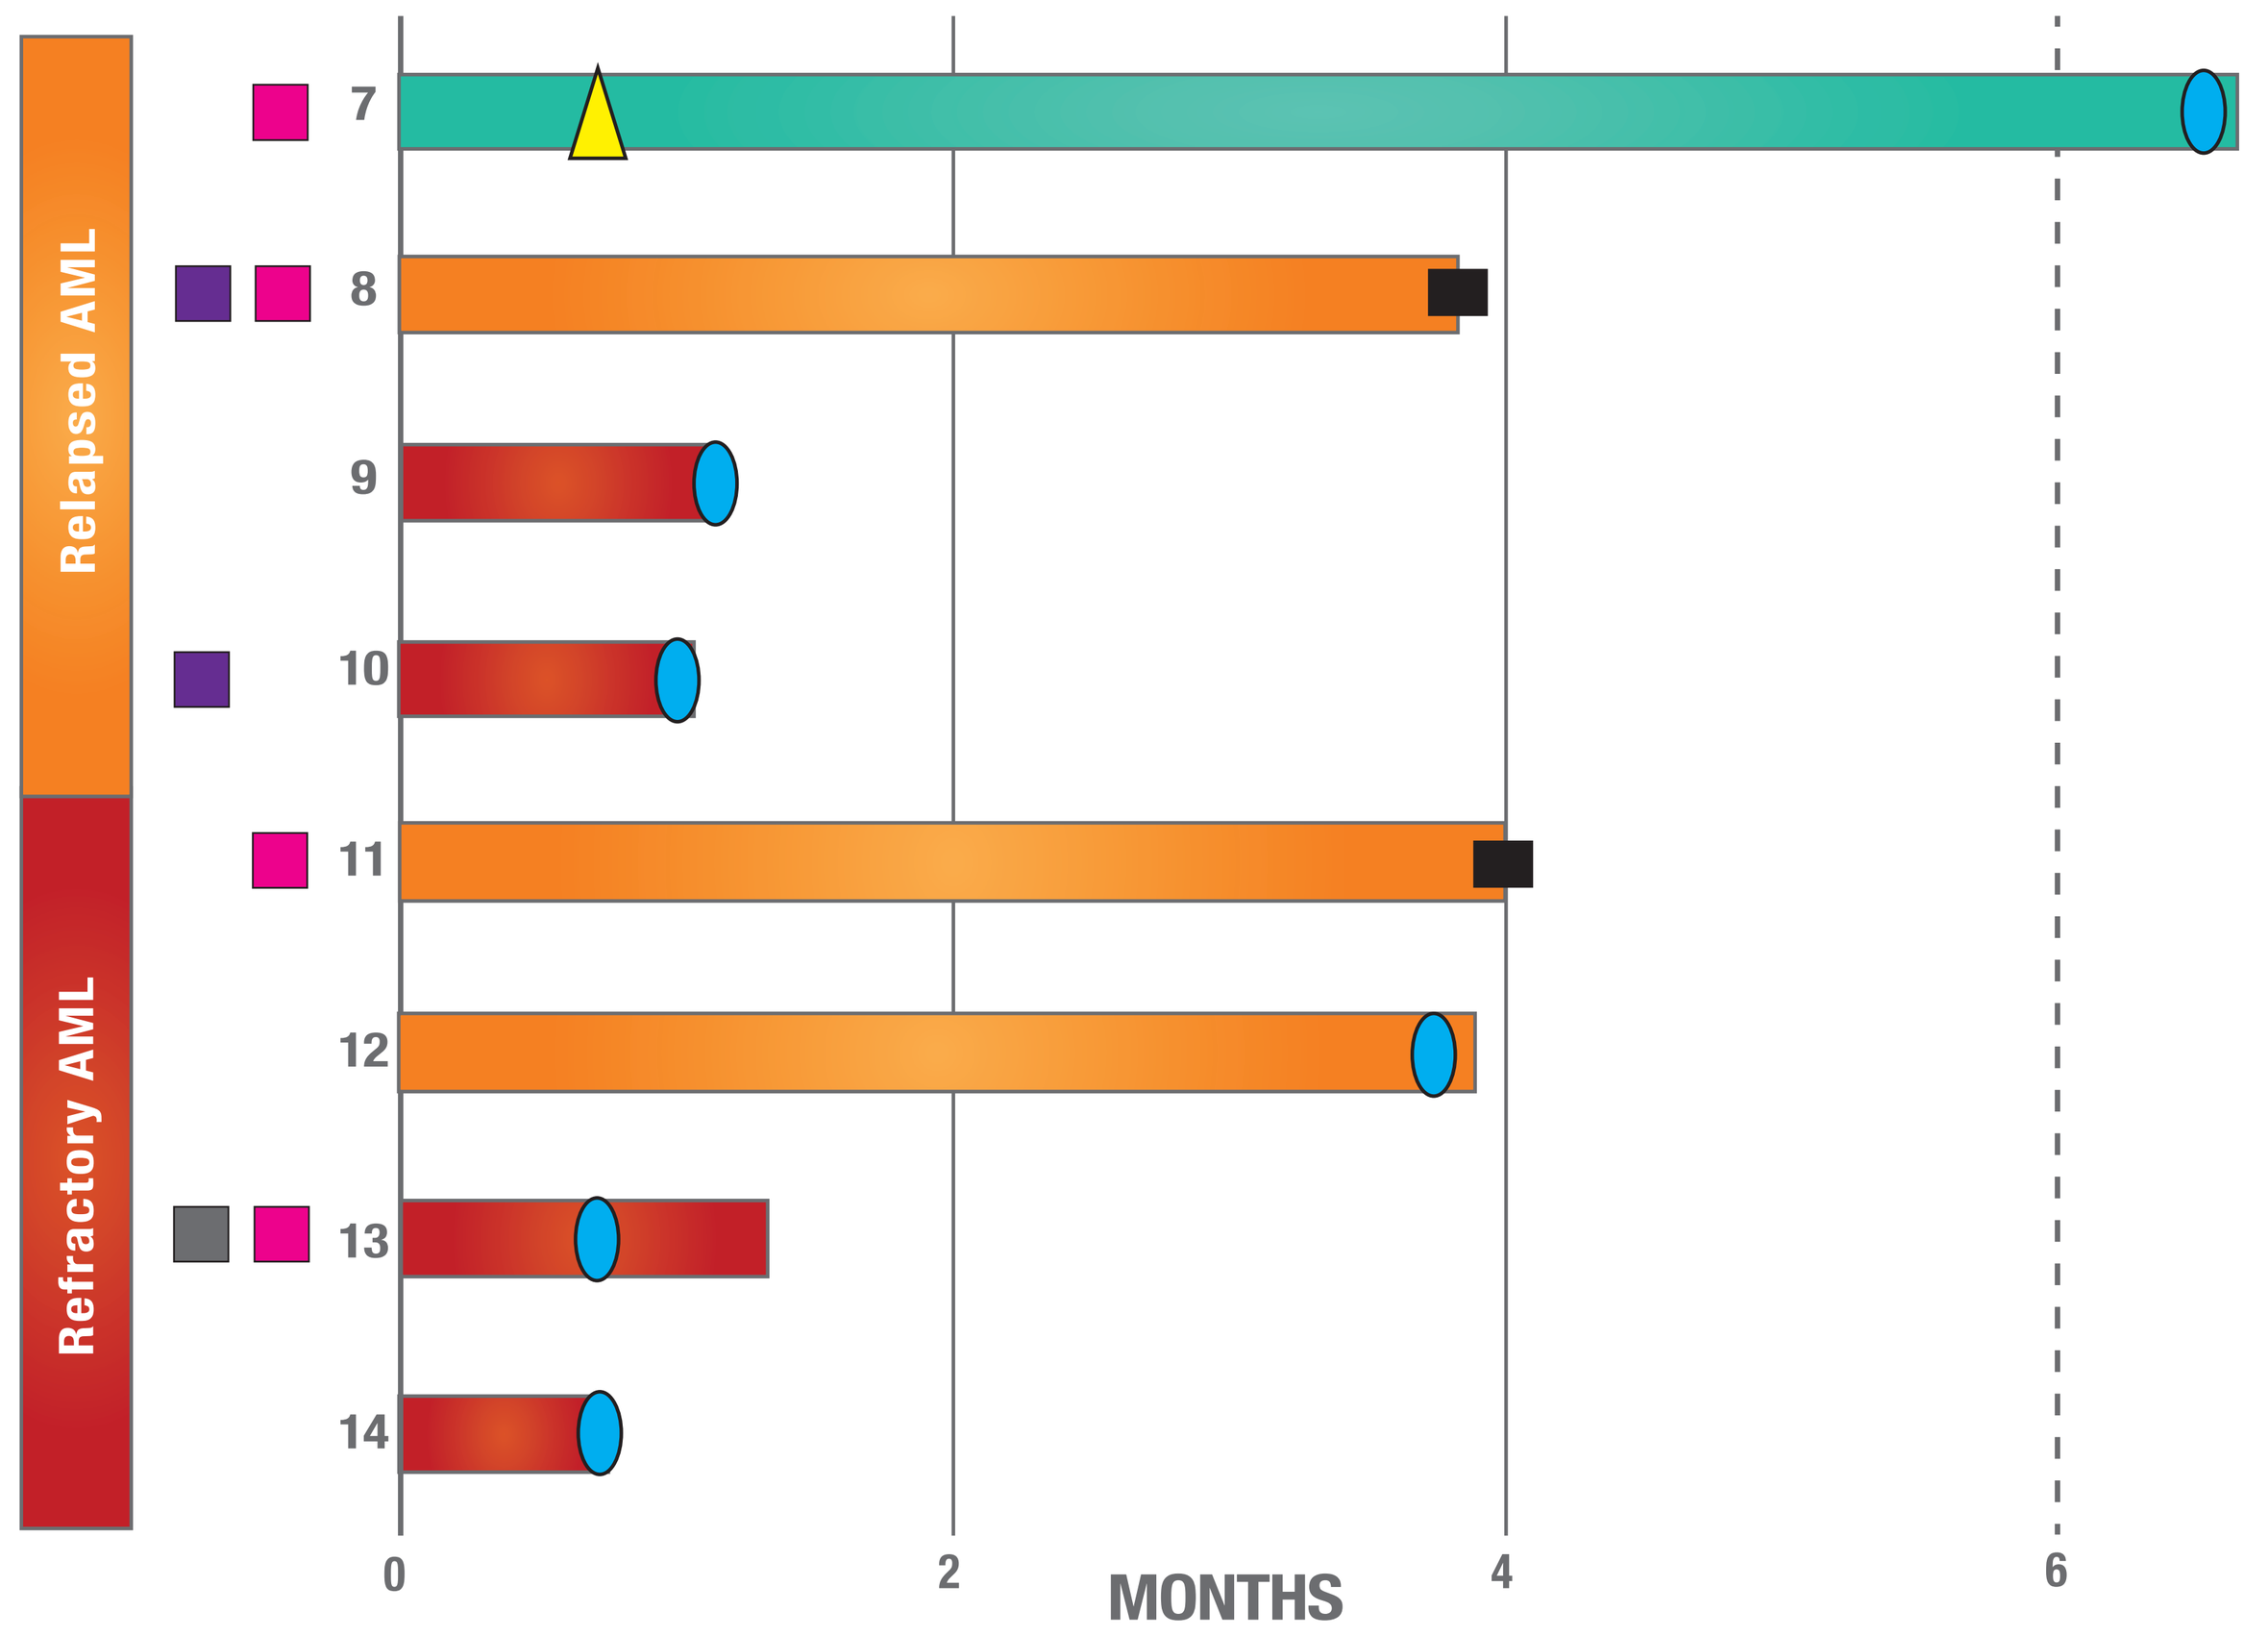
Relapsed AML
Refractory AML
7
8
9
10
11
12
13
14
0
2
4
6
MONTHS

Response Comparison to Current LDAC Combination Therapies

- All patients are newly-diagnosed (first-line) AML
- Data for LDAC combination treatments that are not LDAC + bemceninib are from recently published studies (see references)
- *mDOR not reached: 100% of CR patients are ongoing
- **OS data not yet reported

Combination Therapy	n	CR/CRi	CR/CRi rate (%)	Median DOR (months)	Median OS (months)
LDAC + bemcentinib	6	3	50%	>9.9*	NA**
LDAC + glasdegib[13]	88	24	27%	9.9	8.8
LDAC + venetoclax[14]	82	44	54%	8.1	10.1

Event-free Survival: Newly-Diagnosed Patients

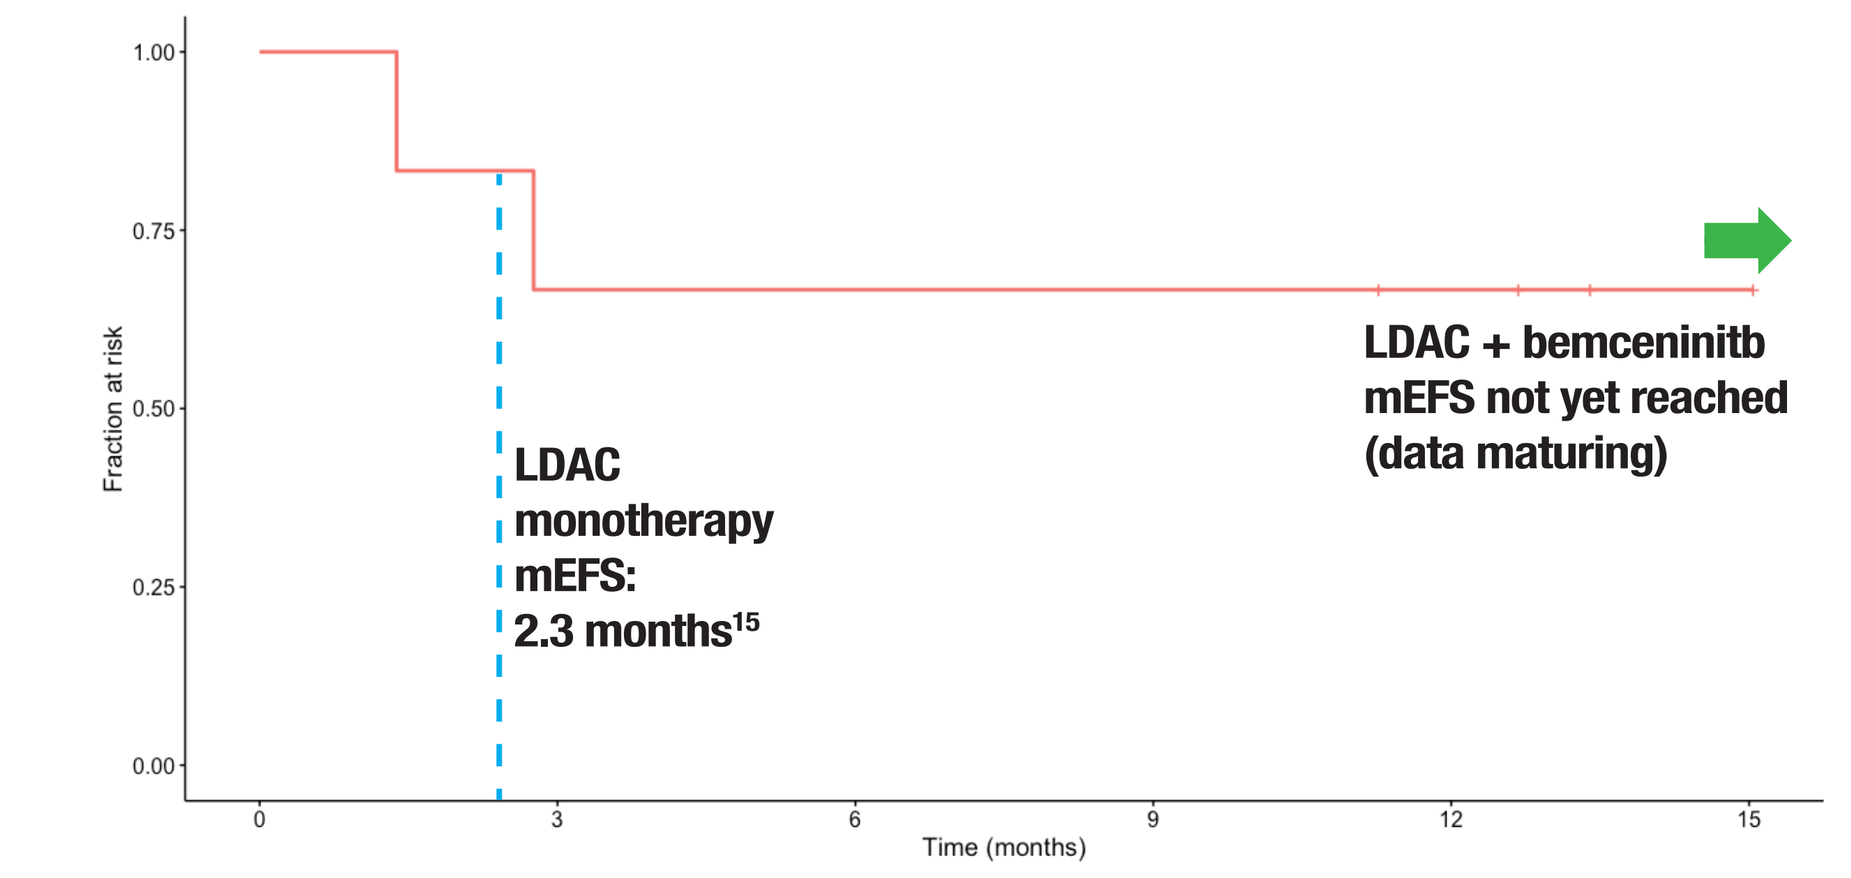
1.00
0.75
0.50
0.25
0.00
Fraction at risk
0
3
6
9
12
15
Time (months)
LDAC monotherapy mEFS: 2.3 months[15]
LDAC + bemceninitb mEFS not yet reached (data maturing)

Patient 4 Case Study: Treatment with bemcentinib beyond progression leads to durable AML control

- 76 year-old male patient with newly-diagnosed secondary AML
- FAB classification of M7: acute megakaryoblastic leukemia (AMKL, only 1% of adult AML, poor prognosis)
- Intermediate cytogenetic risk
- Patient is ongoing

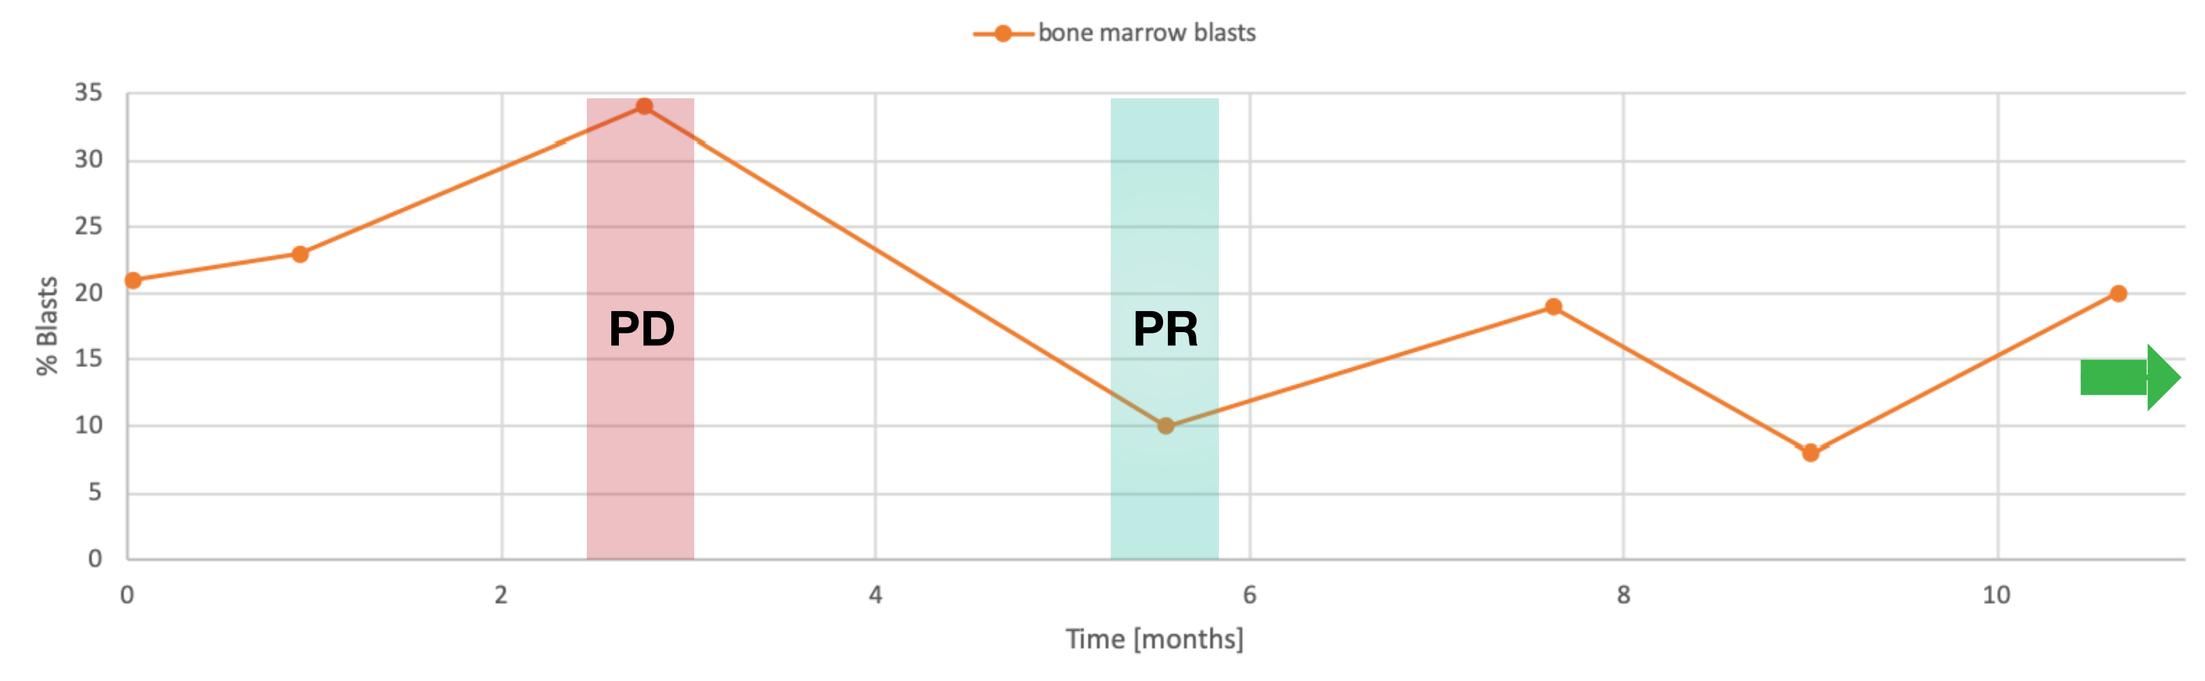
bone marrow blasts
35
30
25
20
15
10
5
0
% Blasts
PD
PR
0
2
4
6
8
10
Time [months]

Biomarkers

- Serum samples taken at C1D1 and C2D1 were analyzed for a panel of >250 circulating biomarker proteins including soluble Axl (sAxl)
- As previously reported[12], pretreatment sAXL levels are associated with response. 6/7 patients with clinical benefit (CR/CRi/PR/SD) had sAXL levels below the previously reported cut-off (ASH, 2018).
- Differential expression analysis of C1D1 (pretreatment) serum samples identified biomarker candidate BGBM033 to be significantly associated with response to bemcentinib. BGBM033 levels were significantly lower in both responder and SD patients in both the bemcentinib-LDAC combination and bemcentinib monotherapy cohorts in this study.
- Notably, low pretreatment levels of BGBM033 were also associated with response to bemcentinib treatment in lung cancer
- Data is still maturing and transcriptional analysis of PBMC and bone marrow samples is ongoing

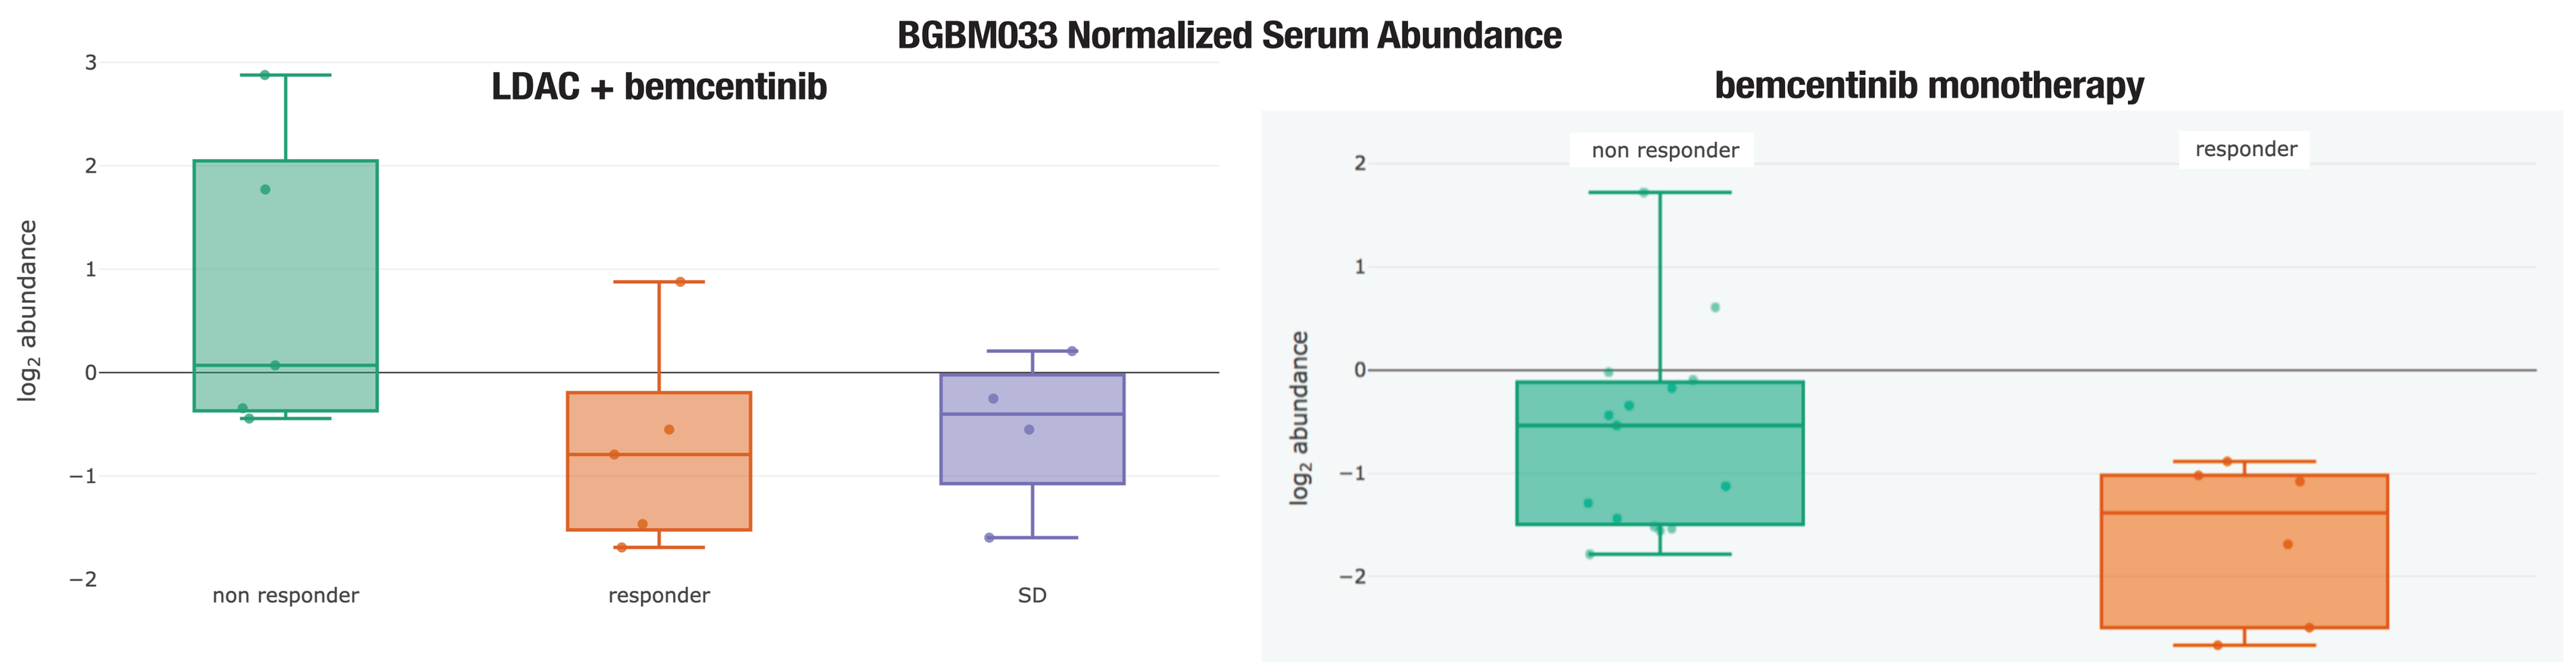
BGBM033 Normalized Serum Abundance
LDAC + bemcentinib
bemcentinib monotherapy
log2 abundance
3
2
1
0
−1
−2
non responder
responder
SD

References

[1] **Cheson, et al.** Journal of Clinical Oncology (2003). Revised recommendations of the International Working Group for Diagnosis, Standardization of Response Criteria, Treatment Outcomes, and Reporting Standards for Therapeutic Trials in Acute Myeloid Leukemia.
[2] **Linger et al.** Adv Cancer Res. (2008) Biologic Functions, Signaling, and Potential Therapeutic Targeting in Human Cancer
[3] **Davidsen, et al.** Springer Publishing (2017). The Role of Axl Receptor Tyrosine Kinase in Tumor Cell Plasticity and Therapy Resistance.
[4] **Zhange et al.** Onco Targets Ther. (2018) The prognostic role of Gas6/Axl axis in solid malignancies: a meta-analysis and literature review
[5] **Gjerdrum et al.** Proc Natl Acad Sci U S A. (2010) Axl is an essential epithelial-to-mesenchymal transition-induced regulator of breast cancer metastasis and patient survival.
[6] **Rochlitz, et al.** Leukemia (1999). Axl expression is associated with adverse prognosis and with expression of Bcl-2 and CD34 in de novo acute myeloid leukemia (AML)
[7] **Ben-Batalla, et al.** Blood (2013). Axl, a prognostic and therapeutic target in acute myeloid leukemia mediates paracrine crosstalk of leukemia cells with bone marrow stroma
[8] **Ludwig et al.** Cancer Res. (2018) Small molecule inhibition of Axl targets tumor immune suppression and enhances chemotherapy in pancreatic cancer
[9] **Byers et al.** EORTC (2016). A Phase I/II and pharmacokinetic study of BGB324, a slective AXL inhibitor as monotherapy and combination with erlotinib in patients with advanced Non-Small Cell Lung Cancer (NSCLC)
[10] **Davidsen et al.** AACR (2018). Bemcentinib (BGB324) a selective small molecule inhibitor of receptor tyrosine kinase AXL, targets tumor immune suppression and enhances immune checkpoint inhibitor efficacy
[11] **Daver et al.** Canc Disc (2019). Efficacy, Safety, and Biomarkers of Response to Azacitidine and Nivolumab in Relapsed/Refractory Acute Myeloid Leukemia: A Nonrandomized, Open-Label, Phase II Study
[12] **Loges et al.** ASH (2018). Comprehensive Analysis of the Dose Escalation, Expansion and Correlates in the Ph I/II Trial BGBC003 with the Selective Oral AXL Inhibitor Bemcentinib (BGB324) in Relapsed/Refractory AML and MDS.
Historical data from clinical trials used in comparisons: [13] **Wei et al.** ASCO (2019). [14] **Döhner et al.** Blood (2014). [15] **Cortes et al.** Leukemia (2019).

Contact

BerGenBio Ltd.
1 Robert Robinson Ave
Oxford
OX4 4GA
UK
post@bergenbio.com
+47 559 61 159

BerGenBio ASA
Jonas Lies vei 91
5009 Bergen
Norway
www.bergenbio.com
@BGenBio

Sonja Loges
University Hamburg-Eppendorf
Martinistr. 52
2024 Hamburg
Germany
s.loges@uke.de
+49 7410 51538

Conclusions

- **The bemcentinib-LDAC combination showed promising efficacy among elderly newly-diagnosed and relapsed AML patients**
- **The overall response rate and duration surpass historical benchmarks and compare favorably to other LDAC combinations**
- **Bemcentinib is safe and well tolerated in combination with LDAC in elderly AML patients**
- **Durable responses and successful treatment beyond progression could be due to an immune-modulatory effect of bemcentinib**
- **Consistent with results in the bemcentinib monotherapy cohort, pretreatment sAXL levels are associated with clinical benefit from LDAC + bemcentinib**
- **A comprehensive biomarker program comprising Axl expression and signalling analysis in tumour and immune compartments is underway to identify both predictive and pharmacodynamic biomarkers**
- **Further clinical development of this combination is warranted**

This presentation is the intellectual property of the presenter. Contact them at post@bergenbio.com for permission to reprint and/or distribute.

This poster was presented at the ASH Annual Meeting 2019, Orlando, FL, USA (Dec 7-10)