Single cell signaling pharmacodynamics in a phase 1b/2 clinical trial of the AXL inhibitor bemcentinib in acute myeloid leukemia and myelodysplastic syndrome

Hellesøy M[1] , Fagerholt OHE[1], Tislevoll BS[1], Gullaksen SE[1], Forthun R[1], Hovland R[1], Reikvam H[1], Kittang AO[1], Cortes JG[2], Lorens J[3], Micklem D[3], Yule M[3], Loges S[4] and Gjertsen BT[1]

[1]*Haukeland University Hospital and University of Bergen Norway* [2]*MDACC United States* [3]*BerGenBio ASA Norway* [4]*Hamburg-Eppendorf University, Germany*

Oda Helen Eck Fagerholt
Medical student
oda.fagerholt@uib.no

Background

Acute myeloid leukemia (AML) is an aggressive hematological disorder caused by clonal expansion of malignant precursor cells. Inhibition of cell differentiation and accumulation of immature cells, causes suppression of normal bone marrow function (1).
To increase the survival rates of AML patients, there is a need for novel treatment strategies, and due to the heterogeneity of the diseases, individually adapted targeted therapy is an intriguing approach.
AXL is a type III receptor tyrosine kinase. Overexpression of AXL is seen in several cancer forms, including AML. As AXL stimulates pro-survival pathways, expression of AXL indicates poor prognosis (2).

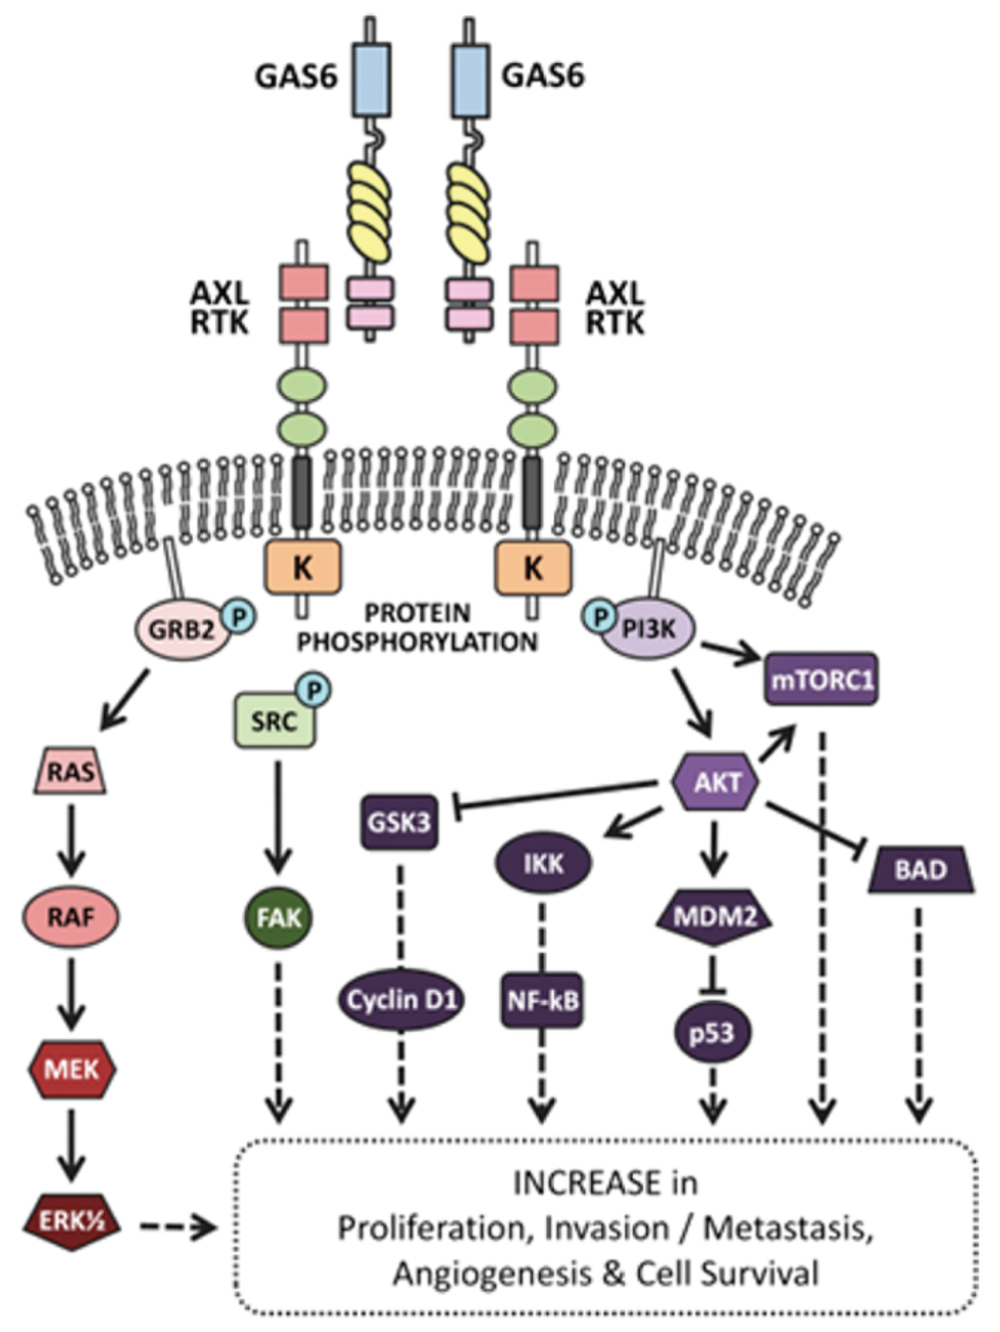
GAS6
GAS6
AXL RTK
AXL RTK
K
K
PROTEIN PHOSPHORYLATION
GRB2
PI3K
mTORC1
SRC
RAS
AKT
GSK3
IKK
BAD
RAF
FAK
MDM2
Cyclin D1
NF-kB
p53
MEK
ERK½
INCREASE in Proliferation, Invasion / Metastasis, Angiogenesis & Cell Survival

BGBC003 Clinical Trial

This phase 1b/2 clinical trial included AML and intermediate/high risk MDS patients. Bemcentinib (BGB324) is a selective potent inhibitor of AXL (2). The drug was orally administered on a daily basis. A total of 24 patients were included in part A. Peripheral blood samples were taken according to a specified schedule, and samples from 6 AML patients were available for single cell profiling.

Methods

Mass cytometry (CyTOF) was used to perform single cell signaling evaluation of treatment response. By using metal-conjugated antibodies, CyTOF allows for simultaneous deep immunophenotyping and evaluation of intracellular signaling responses. Optimum antibody concentrations were determined by titration (3).
The viSNE algorithm was applied for further analysis of the data (4). Surface markers were used for clustering, and blast cells were defined as CD34+ cells.

AML1 CRi | **AML2 Progressive disease** | **AML3 Stable disease** | **AML4 Stable disease** | **AML5 CRi** | **AML6 Progressive disease**

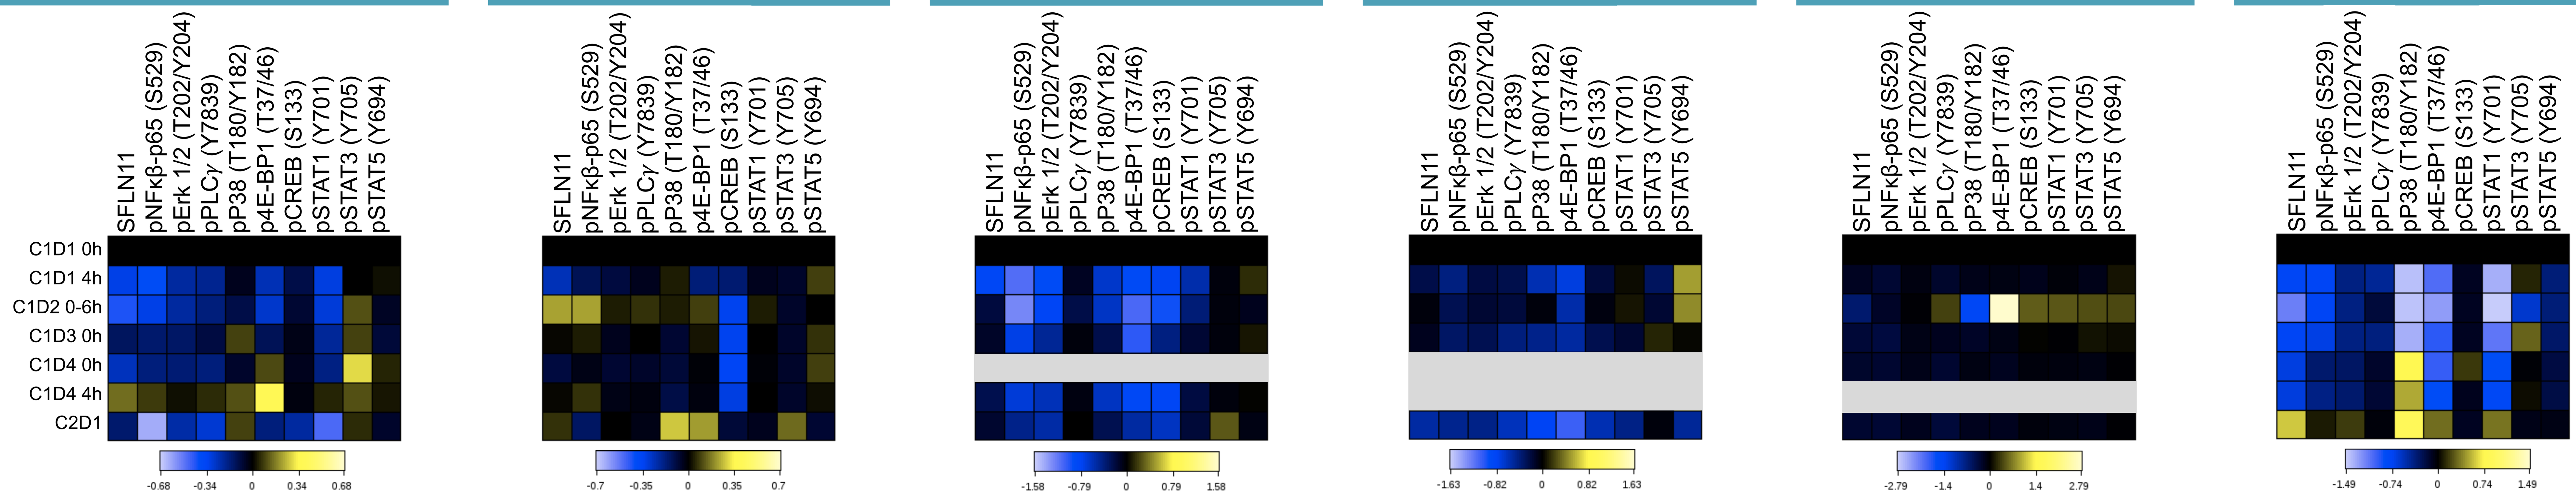
ALL CELLS
SFLN11
pNFκβ-p65 (S529)
pErk 1/2 (T202/Y204)
pPLCγ (Y7839)
pP38 (T180/Y182)
p4E-BP1 (T37/46)
pCREB (S133)
pSTAT1 (Y701)
pSTAT3 (Y705)
pSTAT5 (Y694)
C1D1 0h
C1D1 4h
C1D2 0-6h
C1D3 0h
C1D4 0h
C1D4 4h
C2D1
-0.68 -0.34 0 0.34 0.68
-0.7 -0.35 0 0.35 0.7
-1.58 -0.79 0 0.79 1.58
-1.63 -0.82 0 0.82 1.63
-2.79 -1.4 0 1.4 2.79
-1.49 -0.74 0 0.74 1.49

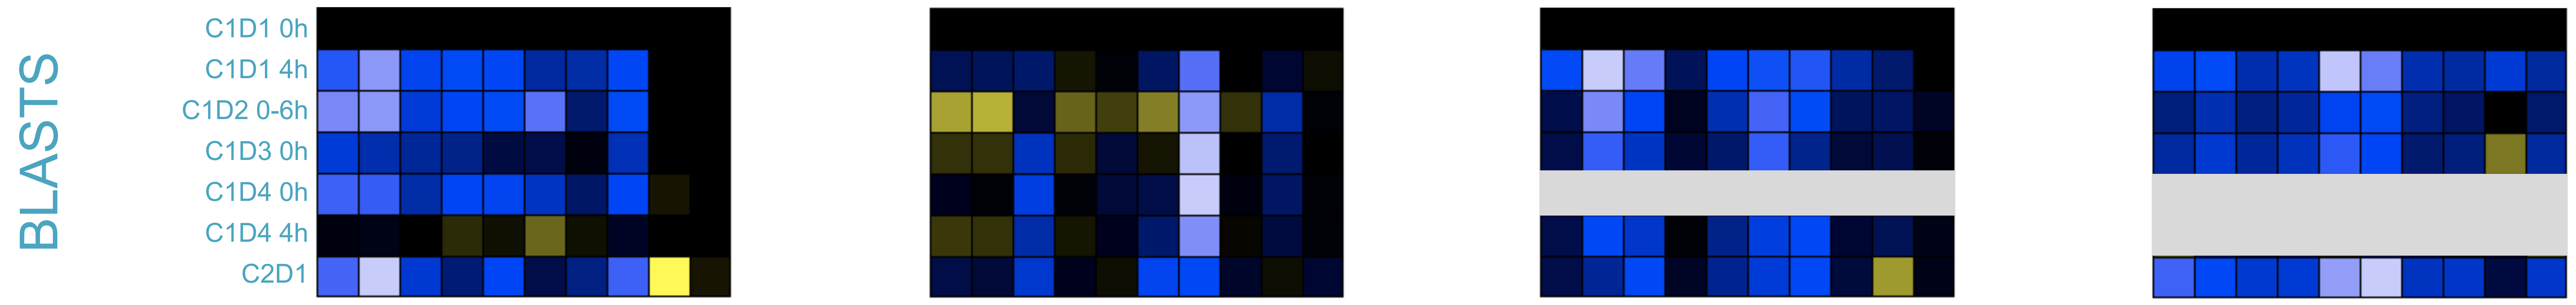
BLASTS
C1D1 0h
C1D1 4h
C1D2 0-6h
C1D3 0h
C1D4 0h
C1D4 4h
C2D1

Heatmaps showing changes of intracellular signaling over the course of therapy. The blood sample taken prior to drug administration is set as reference. Top row (all cells) shows changes in intracellular signaling in the total cell population per blood sample, while bottom row (blasts) shows changes in intracellular signaling in the CD34+ population only. Patient AML5 and AML6 had no defined CD34+ population.

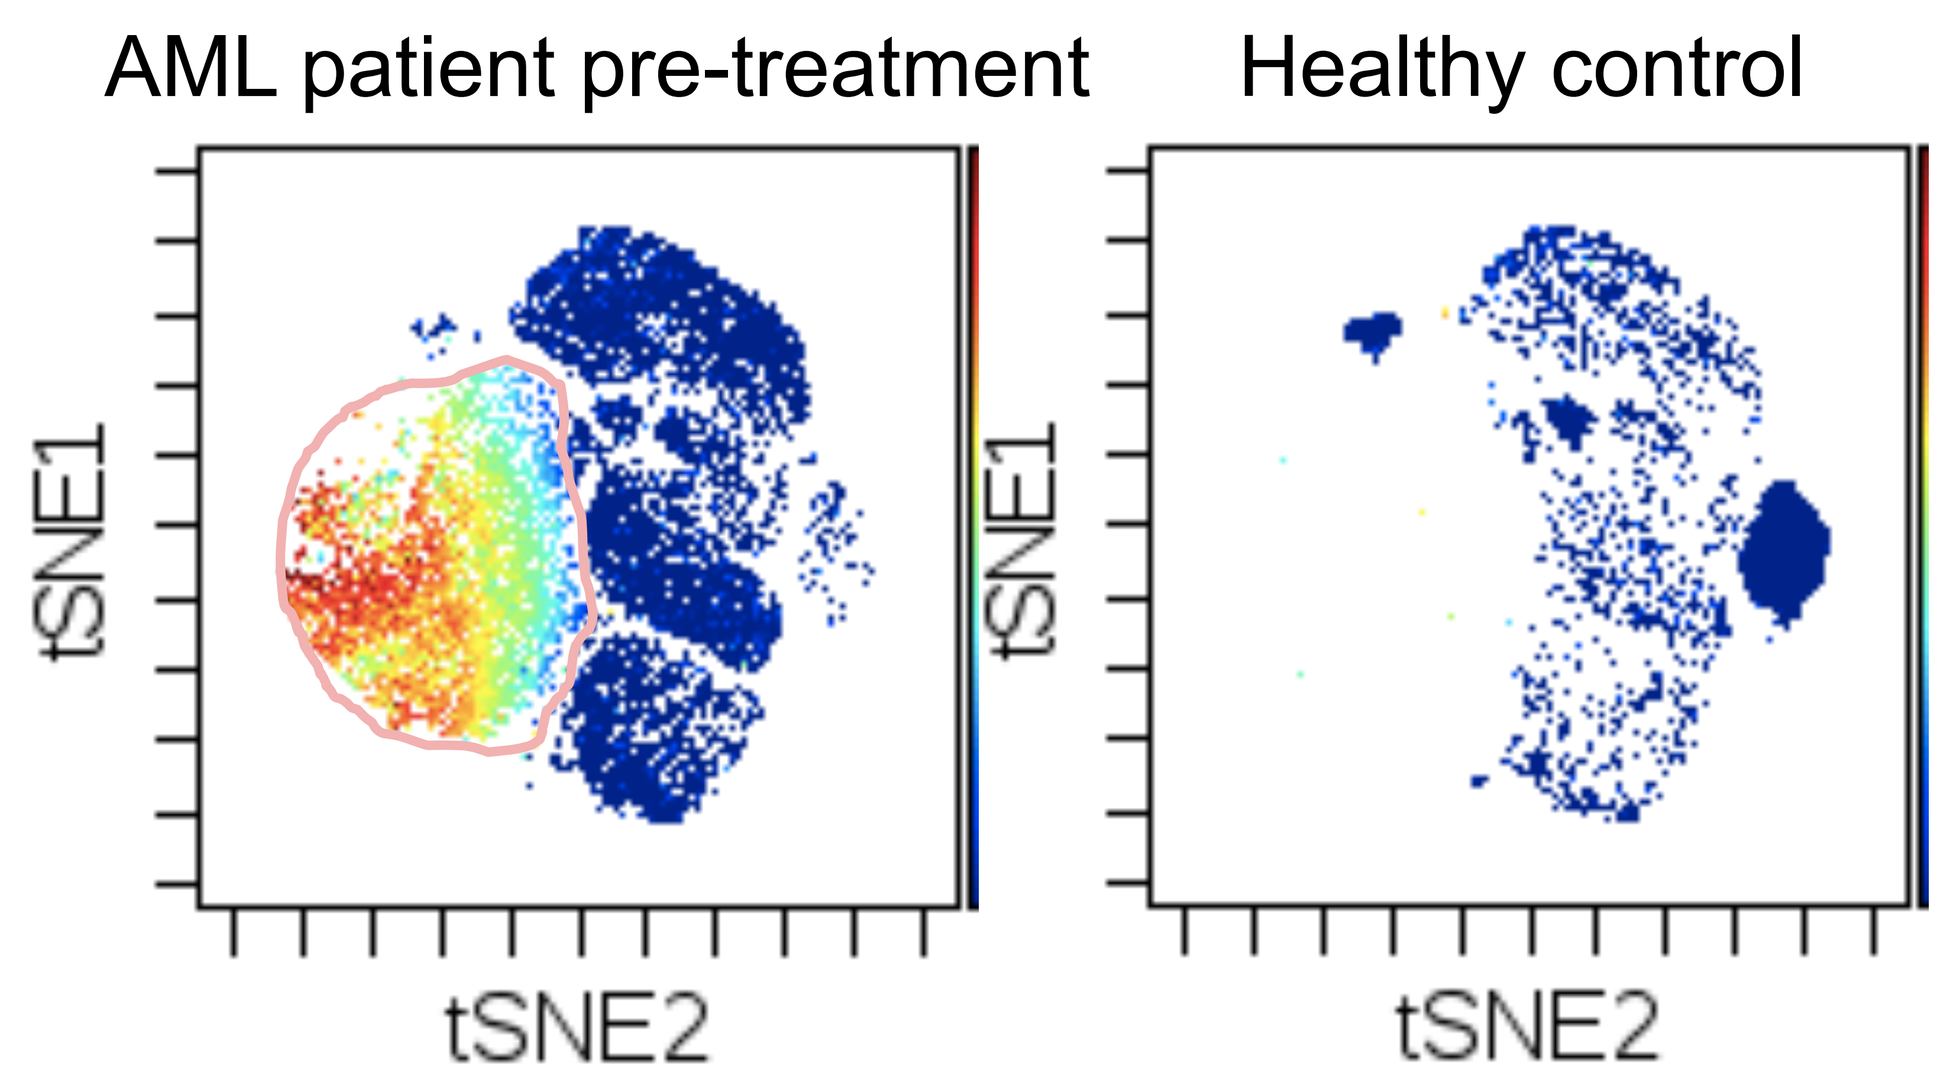
AML patient pre-treatment
Healthy control
tSNE1
tSNE2
tSNE1
tSNE2

The viSNE figure shows the gating strategy of CD34 positive cells exemplified by patient AML 2 (left). The patient blood sample is shown next to a healthy control (right) for comparison.

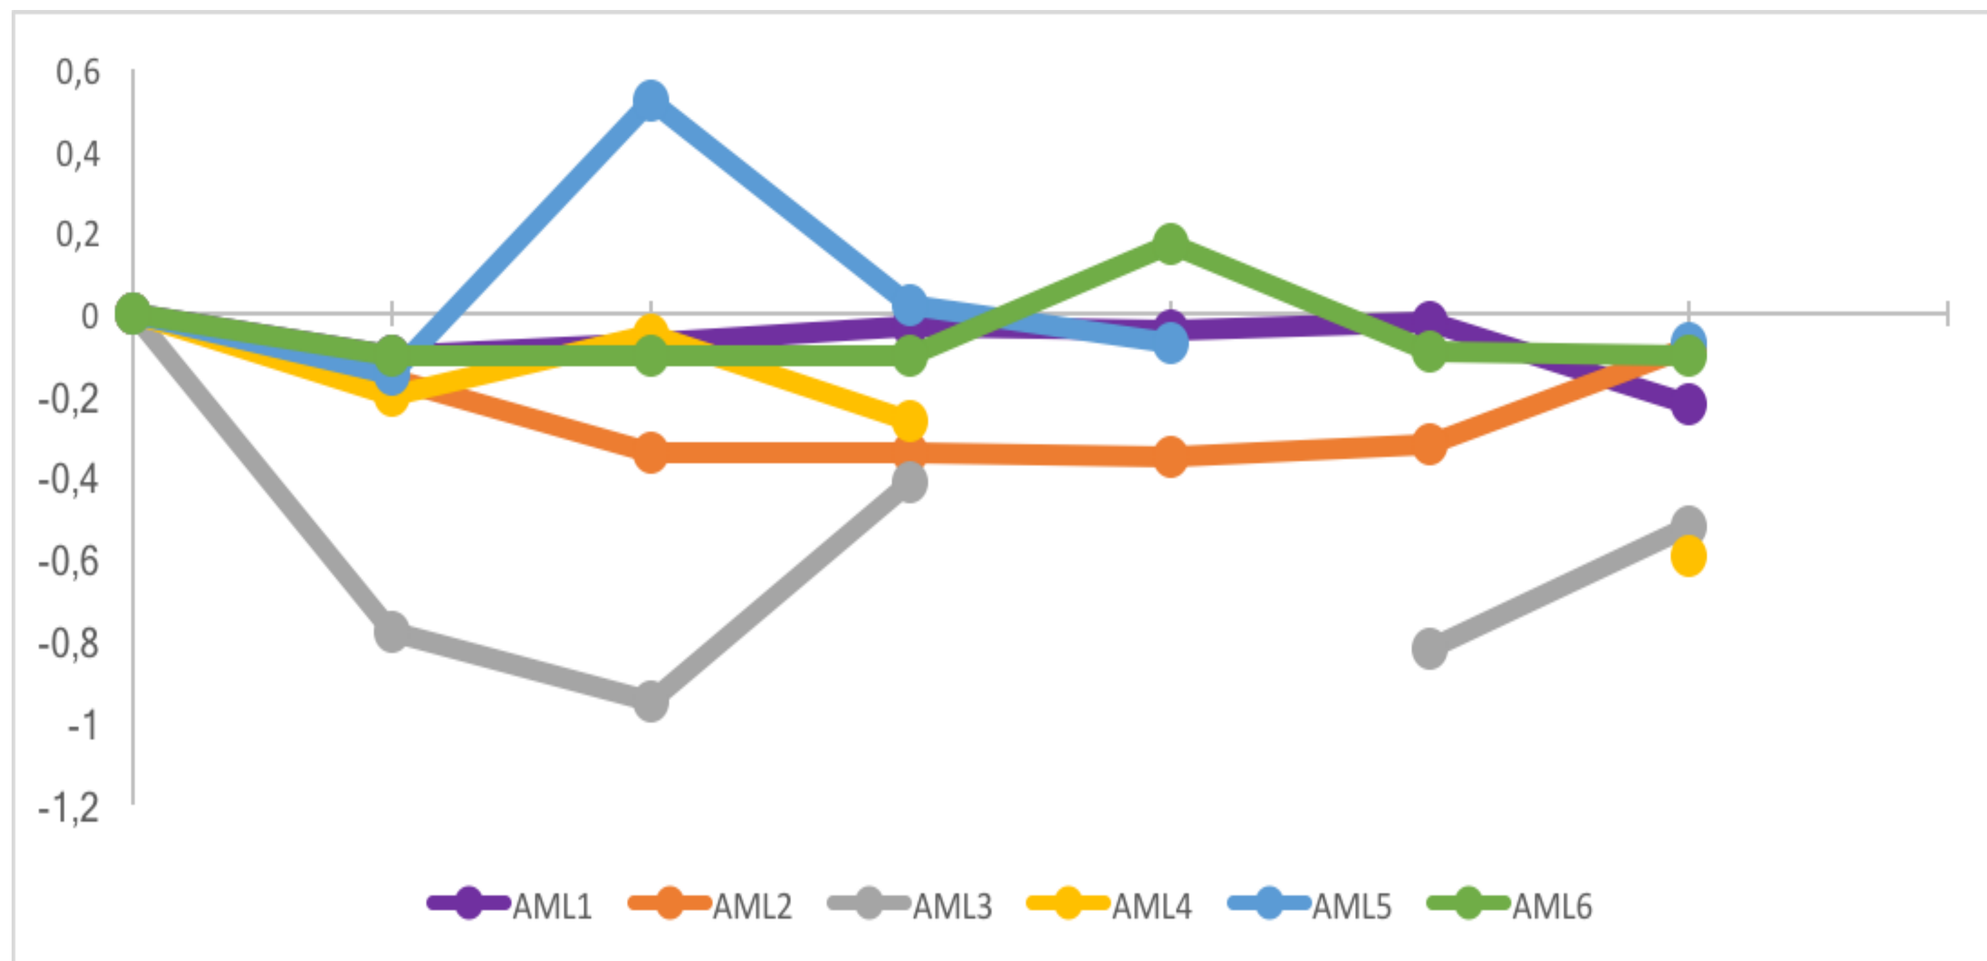
0,6
0,4
0,2
0
-0,2
-0,4
-0,6
-0,8
-1
-1,2
AML1
AML2
AML3
AML4
AML5
AML6

The patients showed heterogeneous changes in intracellular signaling as a response to bemcentinib. However, preliminary results indicates that the two patients without gateable CD34+ population also showed the most prominent pCREB increase.
The graph shows the changes in pCREB signaling in the total cell population of each sample during treatment. The changes are calculated as Arcsinh ratio of medians with pre-administration blood sample as reference.

Conclusion

- By comparing intracellular phospho-signaling changes to pre-treatment sample, we observe changes within 4 hours of drug exposure.
- The patients showed a heterogeneous response to bemcentinib, likely reflecting the heterogenous disease.
- In this small patient cohort and preliminary analysis, we failed to discriminate responders from non-responders by intracellular signaling response.
- Preliminary results indicates that the two patients without gateable CD34+ population also showed the most prominent pCREB increase.

References

1. Short NJ, et al. Lancet. 2018;392(10147):593-606.
2. Holland, SJ et al. Cancer Res. 2010;15;70(4):1544-54.
3. Ben-Batalla I, et al. Blood. 2013;122(14):2443-52.
4. Gullaksen SE, et al. Cytometry A. 2019 Apr 9. doi:10.1002/cyto.a.23751
5. Amir el-AD, et al. Nat Biotechnol. 2013;31(6):545-52.

UNIVERSITY OF BERGEN | Centre for Cancer Biomarkers – Norwegian Centre of Excellence – University of Bergen | HELSE BERGEN Haukeland universitetssjukehus | BerGenBio | KREFTFORENINGEN