A randomized clinical trial of chemotherapy with gemcitabine/cisplatin/nab-paclitaxel with or without the AXL inhibitor bemcentinib (BGB324) for patients with advanced pancreatic cancer.

Muhammad Shaalan Beg[1], Andrew Lowy[2], Peter O' Dwyer[3], Gayle S. Jameson[4], Erkut Hasan Borazanci[4], Hitendra Patel[3], Clare Massey[5], Julia Schoelermann[5], James Lorens[5], Farjana Fattah[1], Kimberli Crane[1], Erin Williams[1], Jatan Clark[6], Daniel Von Hoff[6], Rolf Brekken[1]

(1) Harold C. Simmons Comprehensive Cancer Center, University of Texas Southwestern Medical Center (2) Moores Cancer Center, UC San Diego Health (3) The Presbyterian Medical Center Of Philadelphia (4) HonorHealth Research Institute (5) BerGenBio ASA (6) Translational Genomics Research Institute

UTSouthwestern Medical Center

Abstract #: TPS473

In collaboration with: Penn Medicine Abramson Cancer Center, Stand Up To Cancer, HonorHealth, UC San Diego Medical Center, BerGenBio

Background & objective

RTK AXL drives therapy resistance and negatively regulates innate immunity

The AXL pathway coordinately mediates immune evasion and drug resistance in pancreatic cancer. Systemic AXL inhibition can enhance the efficacy of cancer therapy by blocking tumor cell proliferation, survival and drug resistance associated with epithelial-mesenchymal transition (EMT), and targeting innate immune suppression in the tumor microenvironment.

Bemcentinib (BGB324) is a first in class, selective oral inhibitor of AXL. Our group has shown that bemcentinib therapy, in combination with gemcitabine, improved survival in multiple preclinical models of pancreatic cancer.

AXL signaling in innate immunity *(Graham 2014)*:

- downregulates DC activity
- modulates efferocytosis
- favors immunosuppressive chemokine profile

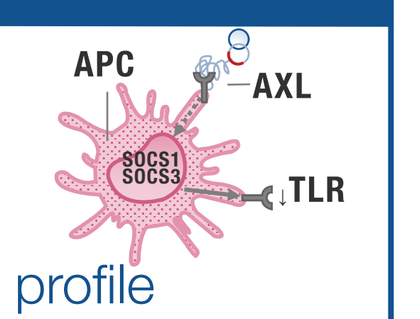
APC
AXL
SOCS1
SOCS3
TLR

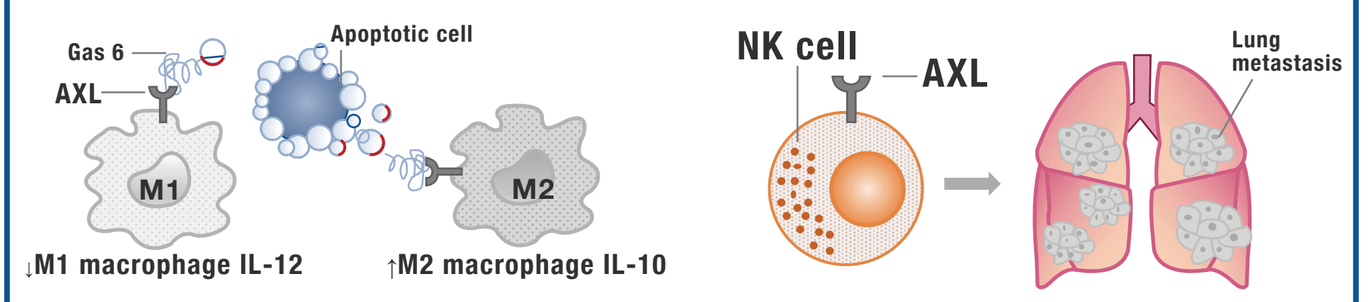
Gas 6
AXL
Apoptotic cell
M1
M2
↓M1 macrophage IL-12
↑M2 macrophage IL-10
NK cell
AXL
Lung metastasis

Bemcentinib - selective, oral small molecule inhibitor of AXL

Bemcentinib is developed in combination with immune-, targeted and chemotherapy in NSCLC, AML/MDS, melanoma and pancreatic cancer

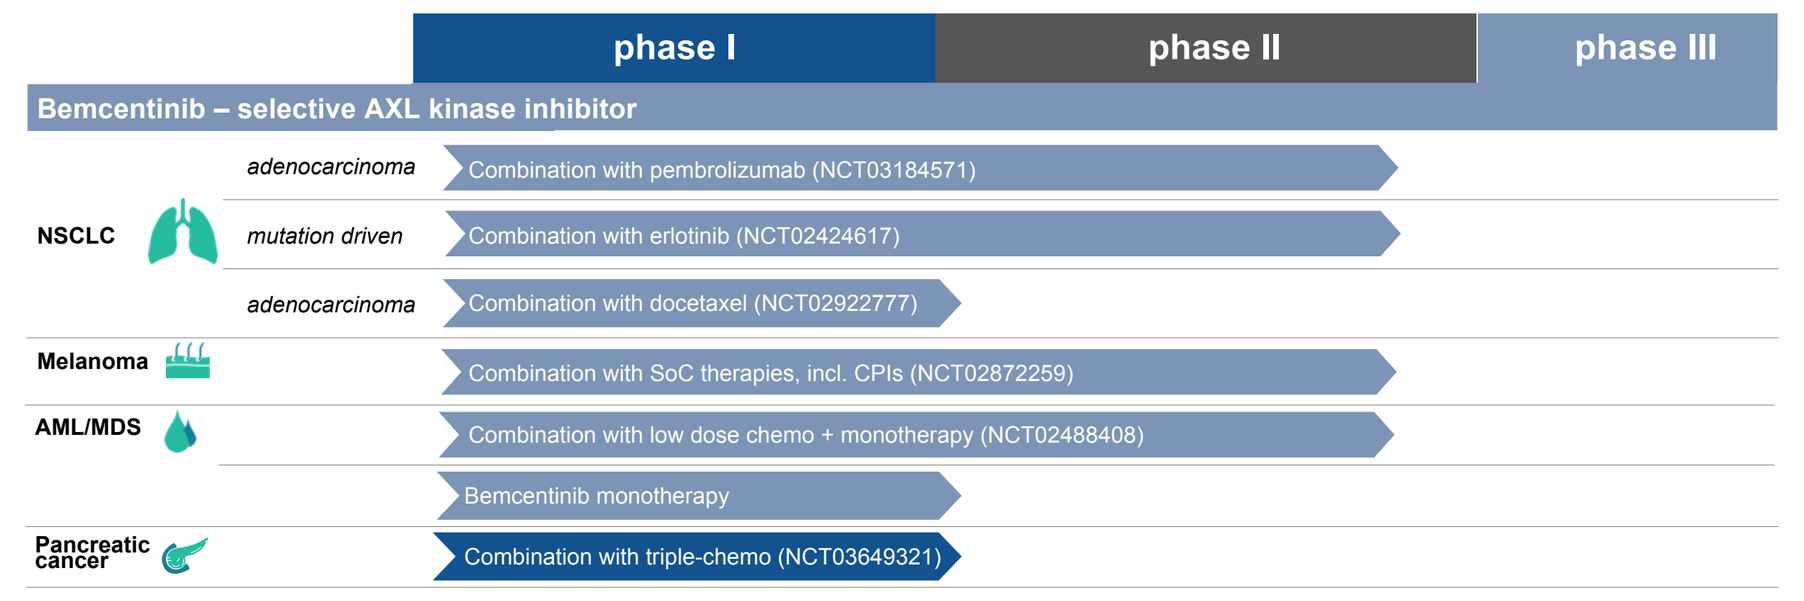
phase I
phase II
phase III
Bemcentinib – selective AXL kinase inhibitor
NSCLC
adenocarcinoma
Combination with pembrolizumab (NCT03184571)
mutation driven
Combination with erlotinib (NCT02424617)
adenocarcinoma
Combination with docetaxel (NCT02922777)
Melanoma
Combination with SoC therapies, incl. CPIs (NCT02872259)
AML/MDS
Combination with low dose chemo + monotherapy (NCT02488408)
Bemcentinib monotherapy
Pancreatic cancer
Combination with triple-chemo (NCT03649321)

Selectivity Profile

Kinase	Kinome Scan (Kd) nM	Fold
Axl	0.4	1
Mer	100	250
Tyro	>1000	>1000

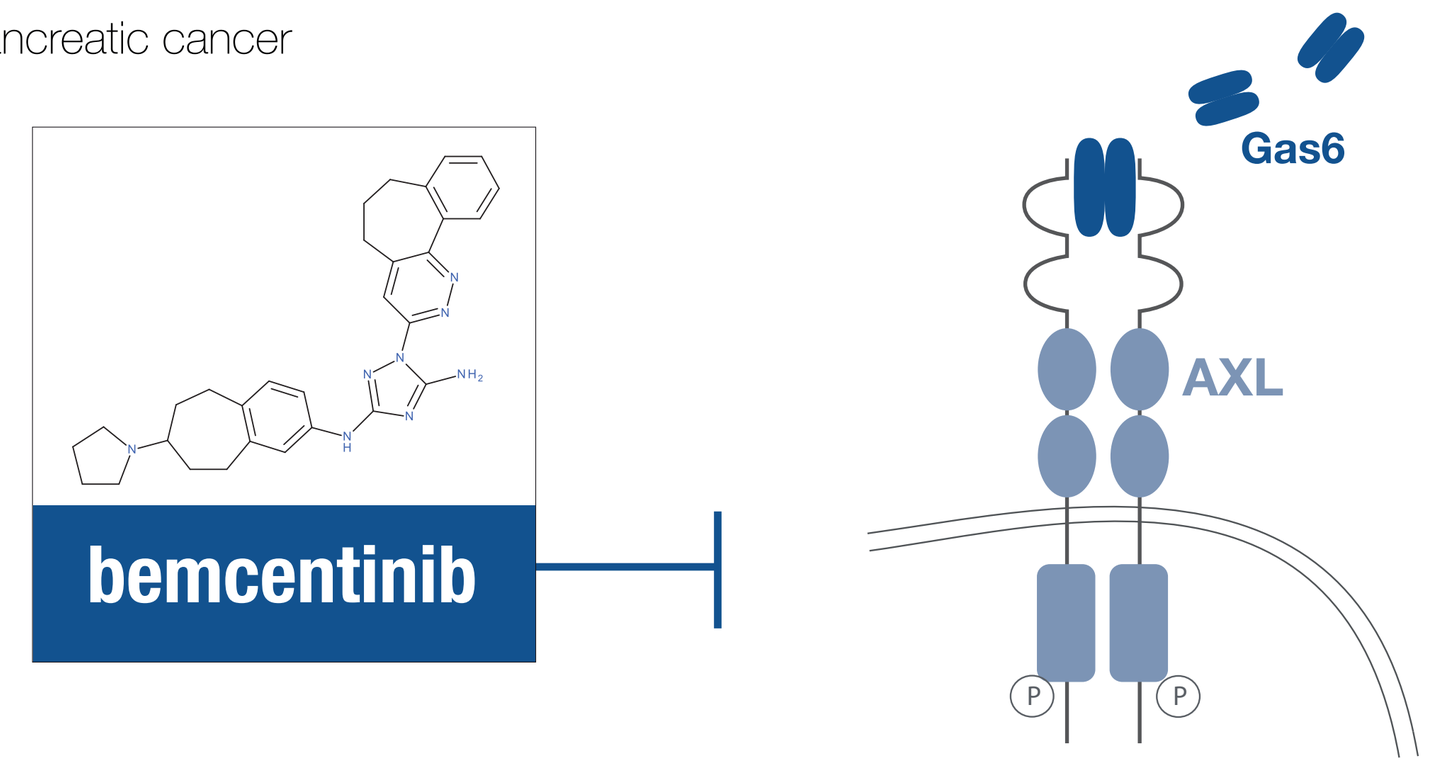
bemcentinib
Gas6
AXL
P
P

Bemcentinib plus chemotherapy have additive effect in models of pancreatic cancers *(Ludwig 2018)*

Bemcentinib targets tumor Immune suppression and enhances chemotherapy in pancreatic cancer

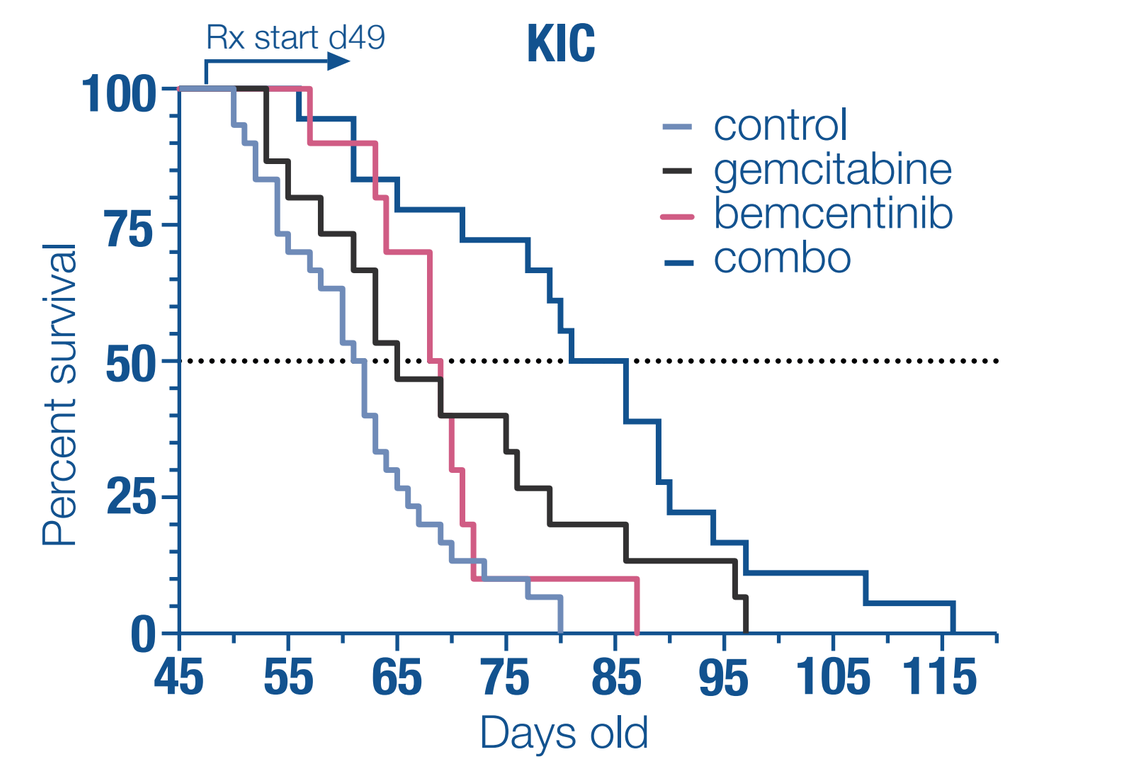
Rx start d49
KIC
control
gemcitabine
bemcentinib
combo
Percent survival
Days old

AXL inhibition in mice with advanced PDAC improves survival. KIC mice were enrolled in a survival study and randomized to vehicle [0.5% (w/w) hydroxypropyl methylcellulose/0.1% (w/w) Tween 80 in water, control, orally BID], bemcentinib (50 mg/kg orally BID), gemcitabine (25 mg/kg i.p. twice a week), and gemcitabine + bemcentinib. Therapy was initiated at day 49 after tumor cell injection and maintained until sacrifice.

Panc265 tumor growth was effectively controlled by combination therapy

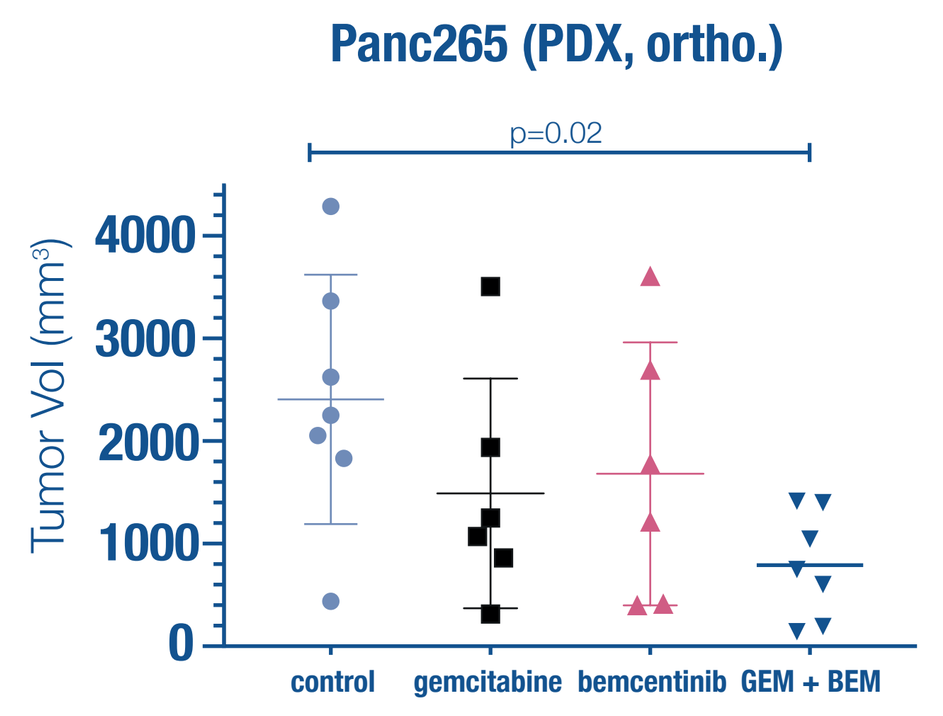
Panc265 (PDX, ortho.)
p=0.02
Tumor Vol (mm³)
control
gemcitabine
bemcentinib
GEM + BEM

Metastases	Spleen	Small Intestine	Kidney	Liver	Lung	Lymph nodes
control (n=7)	7/7	7/7	0/7	7/7	1/7	0/7
gemcitabine (n=6)	6/6	6/6	0/6	2/6	0/6	2/6
bemcentinib (n=6)*	2/6	0/6	0/6	0/6	0/6	0/6
combo (n=7)*	2/7	1/7	0/7	0/7	0/7	0/7

Panc 265 was orthotopically implanted into nu/nu athymic mice. After tumor establishment, mice were treated with vehicle (control, orally BID), bemcentinib (50 mg/kg orally BID), gemcitabine (25 mg/kg i.p. twice a week), or the combination (n = 6–7/group). Total gross metastasis was determined by evaluation of liver, diaphragm, GI lymph nodes, and lung at the time of sacrifice. Bemcentinib alone or in combination with gemcitabine significantly reduced the rate of metastasis (*p=0.001 vs control, Fisher's exact test)

Treatment with bemcentinib alters the immune landscape of PDAC *(Ludwig 2018)*

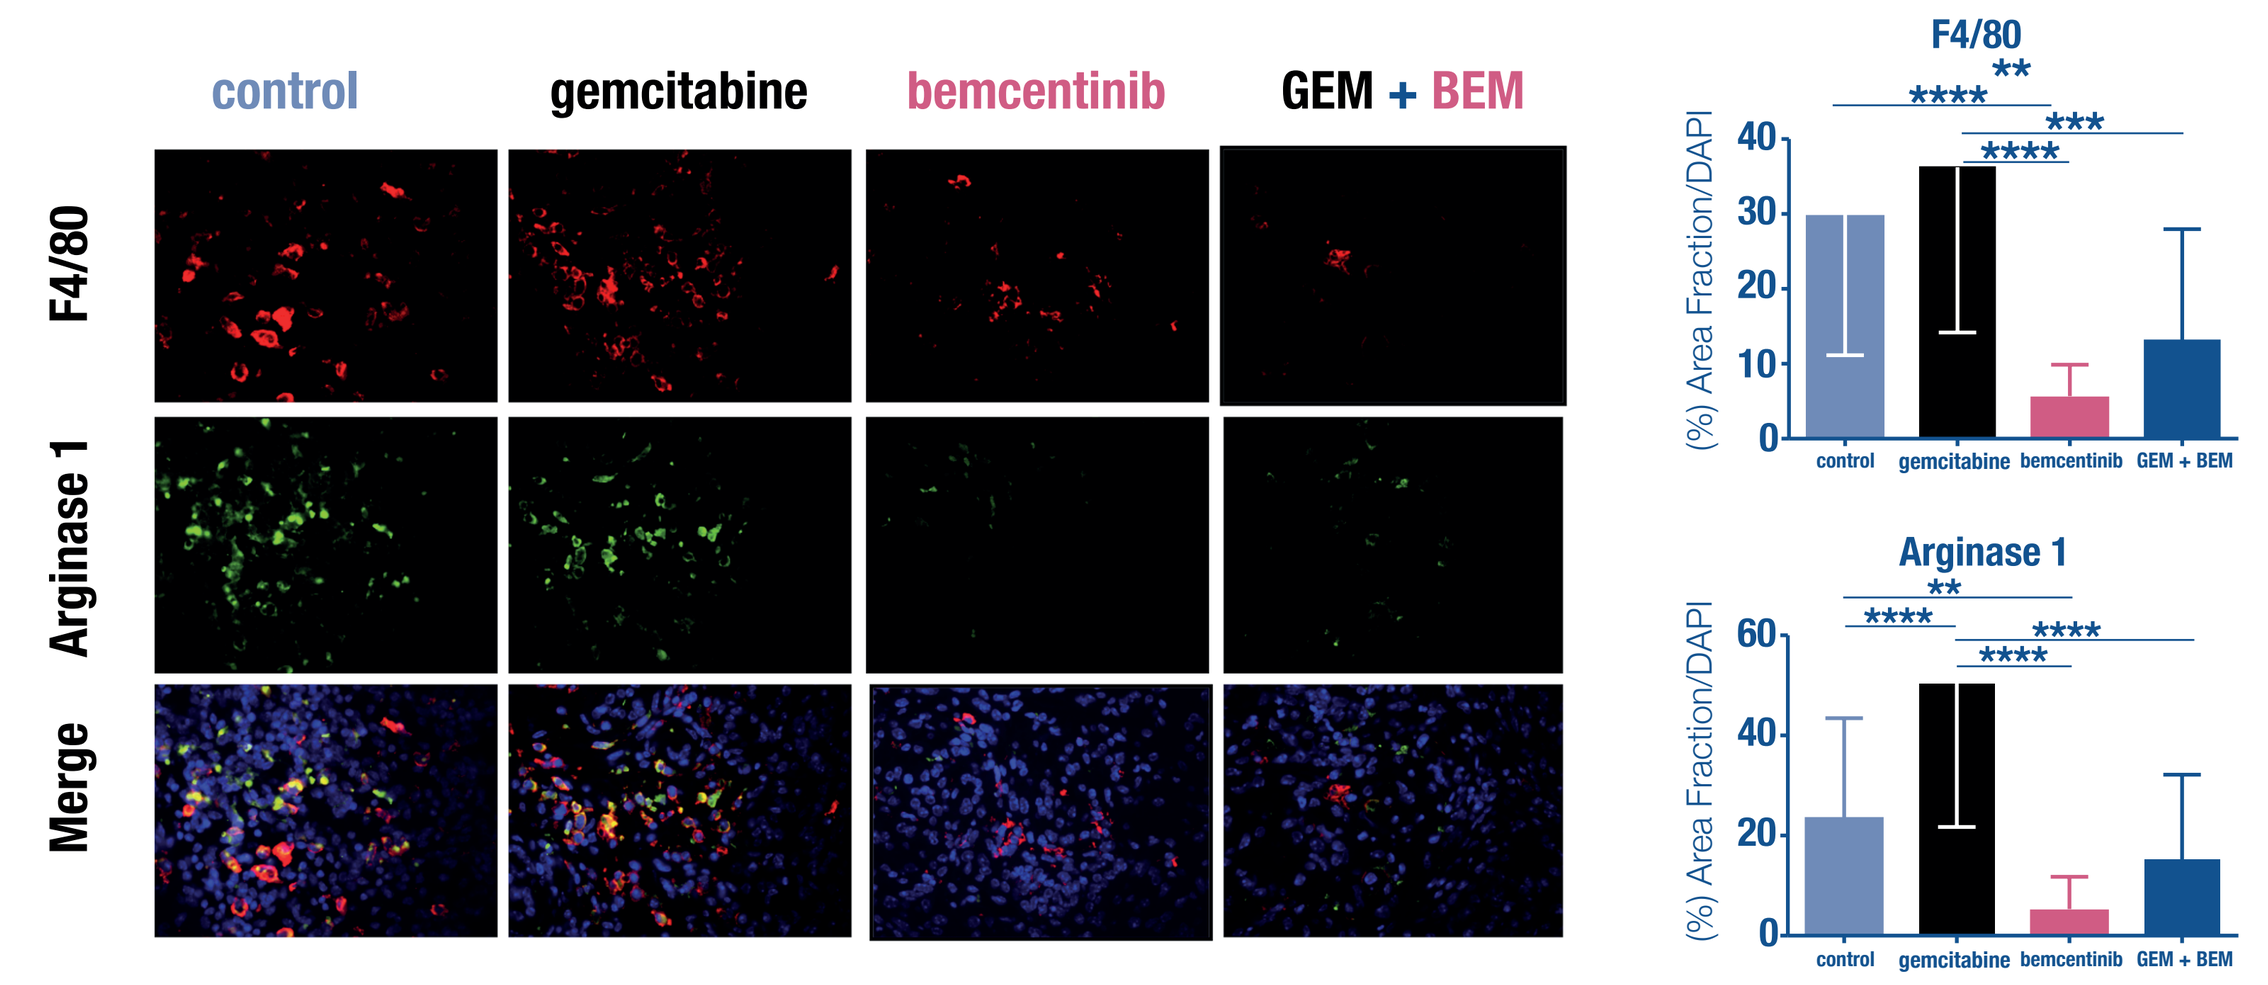
control
gemcitabine
bemcentinib
GEM + BEM
F4/80
Arginase 1
Merge
F4/80
(%) Area Fraction/DAPI
Arginase 1

Tumor tissue from KIC mice treated with bemcentinib +/– gemcitabine were evaluated by immunofluorescence for macrophage (F4/80) and Arginase 1 (Arg1) and counterstained with DAPI. Images were analyzed using Elements software; quantification of a percentage of area fraction is shown. Data are displayed as mean ± SD and represent 5 images per tumor with 4 animals per group analyzed. ** p < 0.01; *** p < 0.005; **** P < 0.001; by ANOVA with Tukey's MCT.

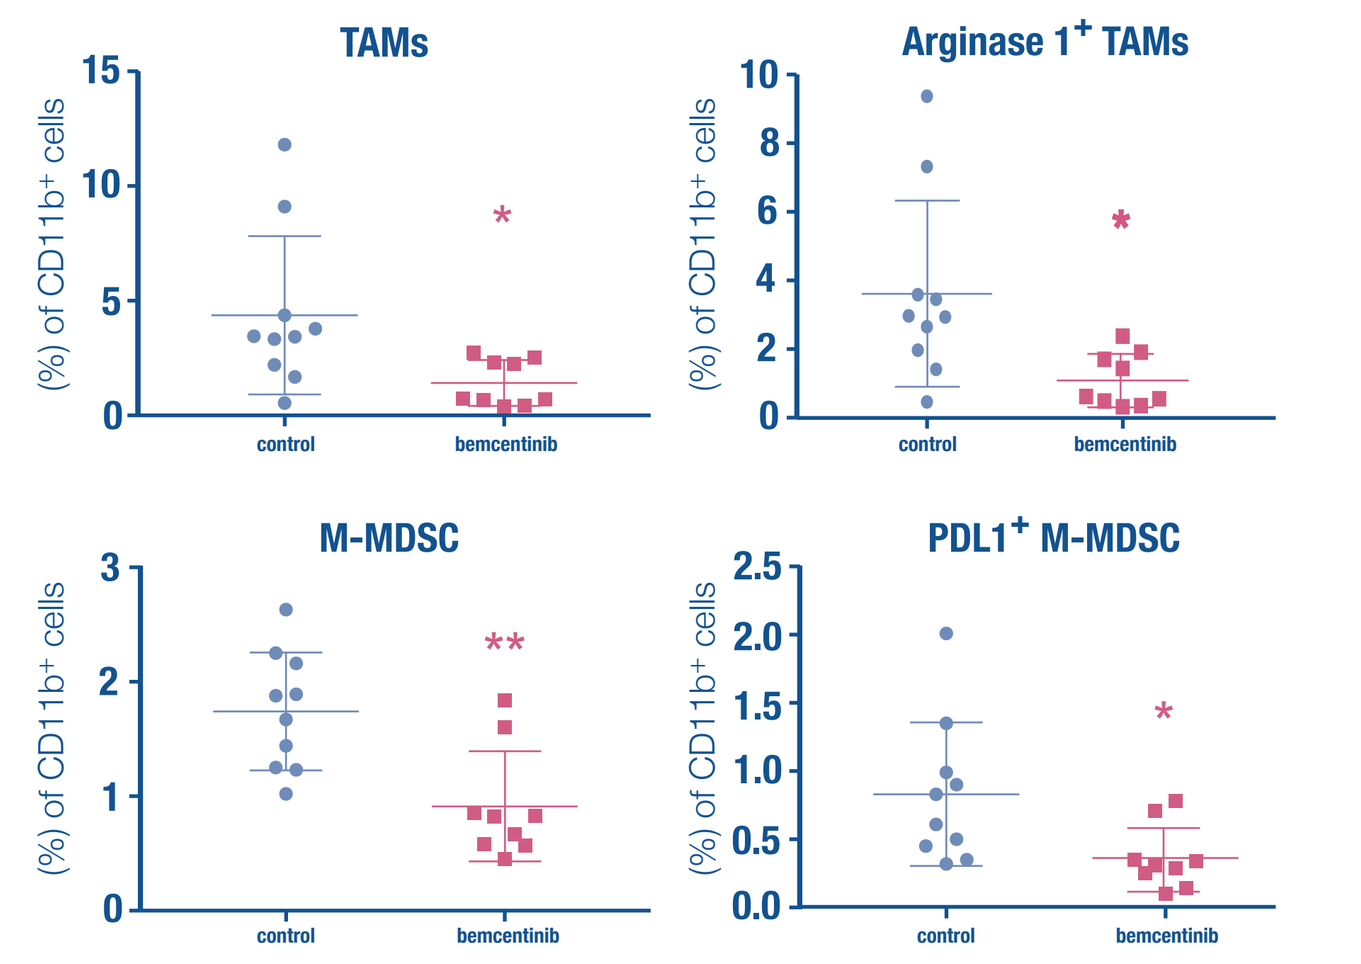
TAMs
Arginase 1+ TAMs
M-MDSC
PDL1+ M-MDSC
(%) of CD11b+ cells
control
bemcentinib

Flow cytometry of KPC-M09 subcutaneous tumors treated with vehicle (control) or bemcentinib (50 mg/kg, PO, BID) for two weeks. Bemcentinib reduced monocytic MDSCs ($CD11b^+ Ly6G^- Ly6C^+$), PD-L1$^+$ M-MDSCs, tumor associated macrophages (TAMs, $CD11b^+ Ly6G^- Ly6C^- F4/80^+ CD11c^+ MHCII^+$) and Arg1$^+$ TAMs. * $p < 0.05$; ** $p < 0.01$ by t-test.

Randomized Ph Ib/II trial of oral, selective AXL inhibitor bemcentinib in combination with chemotherapy

Study Design

Randomized, open-label trial of bemcentinib plus chemotherapy

- Bemcentinib in combination with nab-paclitaxel, gemcitabine and cisplatin (Jameson 2017)
- Previously untreated pancreatic adenocarcinoma that is metastatic or recurrent

Part 1

Safety run in (n=3-12) bemcentinib + chemotherapy

R 1:1

Part 2

Bemcentinib + Chemotherapy (n=31)

- Bemcentinib 200 mg oral daily
- Nab-paclitaxel 125 mg/m2 Day 1/8 every 21 days
- Gemcitabine 1000 mg/m2 Day 1/8 every 21 days
- Cisplatin 25 mg/m2 Day 1/8 every 21 days

Chemotherapy alone (n=31)

- Nab-paclitaxel 125 mg/m2 Day 1/8 every 21 days
- Gemcitabine 1000 mg/m2 Day 1/8 every 21 days
- Cisplatin 25 mg/m2 Day 1/8 every 21 days

Study Objectives

Primary objectives:

- Determine complete response rate of bemcentinib plus triple chemotherapy

Secondary objectives:

- To determine overall response rate (ORR), median progression free survival (PFS), 1-year and 2-year overall survival (OS) rate of bemcentinib plus triple chemotherapy
- Safety and tolerability of bemcentinib plus chemotherapy in patients with metastatic pancreatic adenocarcinoma
- Determine impact of therapy on Quality of Life (QoL)

Exploratory objectives:

- Compare the effect of treatment, with chemotherapy alone vs chemotherapy + AXL inhibitor bemcentinib on changes in tissue and blood biomarkers
- Correlate changes in tissue and blood biomarkers with clinical outcomes (ORR)

Major Inclusion & Exclusion Criteria

Key inclusion criteria:

- Patients must have a histologically or cytologically confirmed pancreatic adenocarcinoma that is metastatic or recurrent
- No prior systemic therapy for metastatic or recurrent disease
 a. Prior adjuvant gemcitabine, if completed more than 12 months prior to date of enrollment, is acceptable
 b. Radiosensitizing chemotherapy, if completed at least 4 weeks from date of enrollment, is acceptable.
- Measurable disease is required per RECIST 1.1 criteria
- Age 18-70 years at the time of enrollment
- ECOG performance status 0 or 1

Key exclusion criteria:

- Patients with known untreated brain metastases
- Has a known additional malignancy that is progressing or requires active treatment
- Known active infection with human immunodeficiency virus (HIV), hepatitis B or C viruses
- Treatment with any medication which is metabolized by CYP3A4 and has a narrow therapeutic index
- Major surgery within 4 weeks prior to date of enrollment; excluding skin biopsies and procedures for insertion of central venous access devices
- Full inc/exc criteria are specified in the protocol

Study Sites

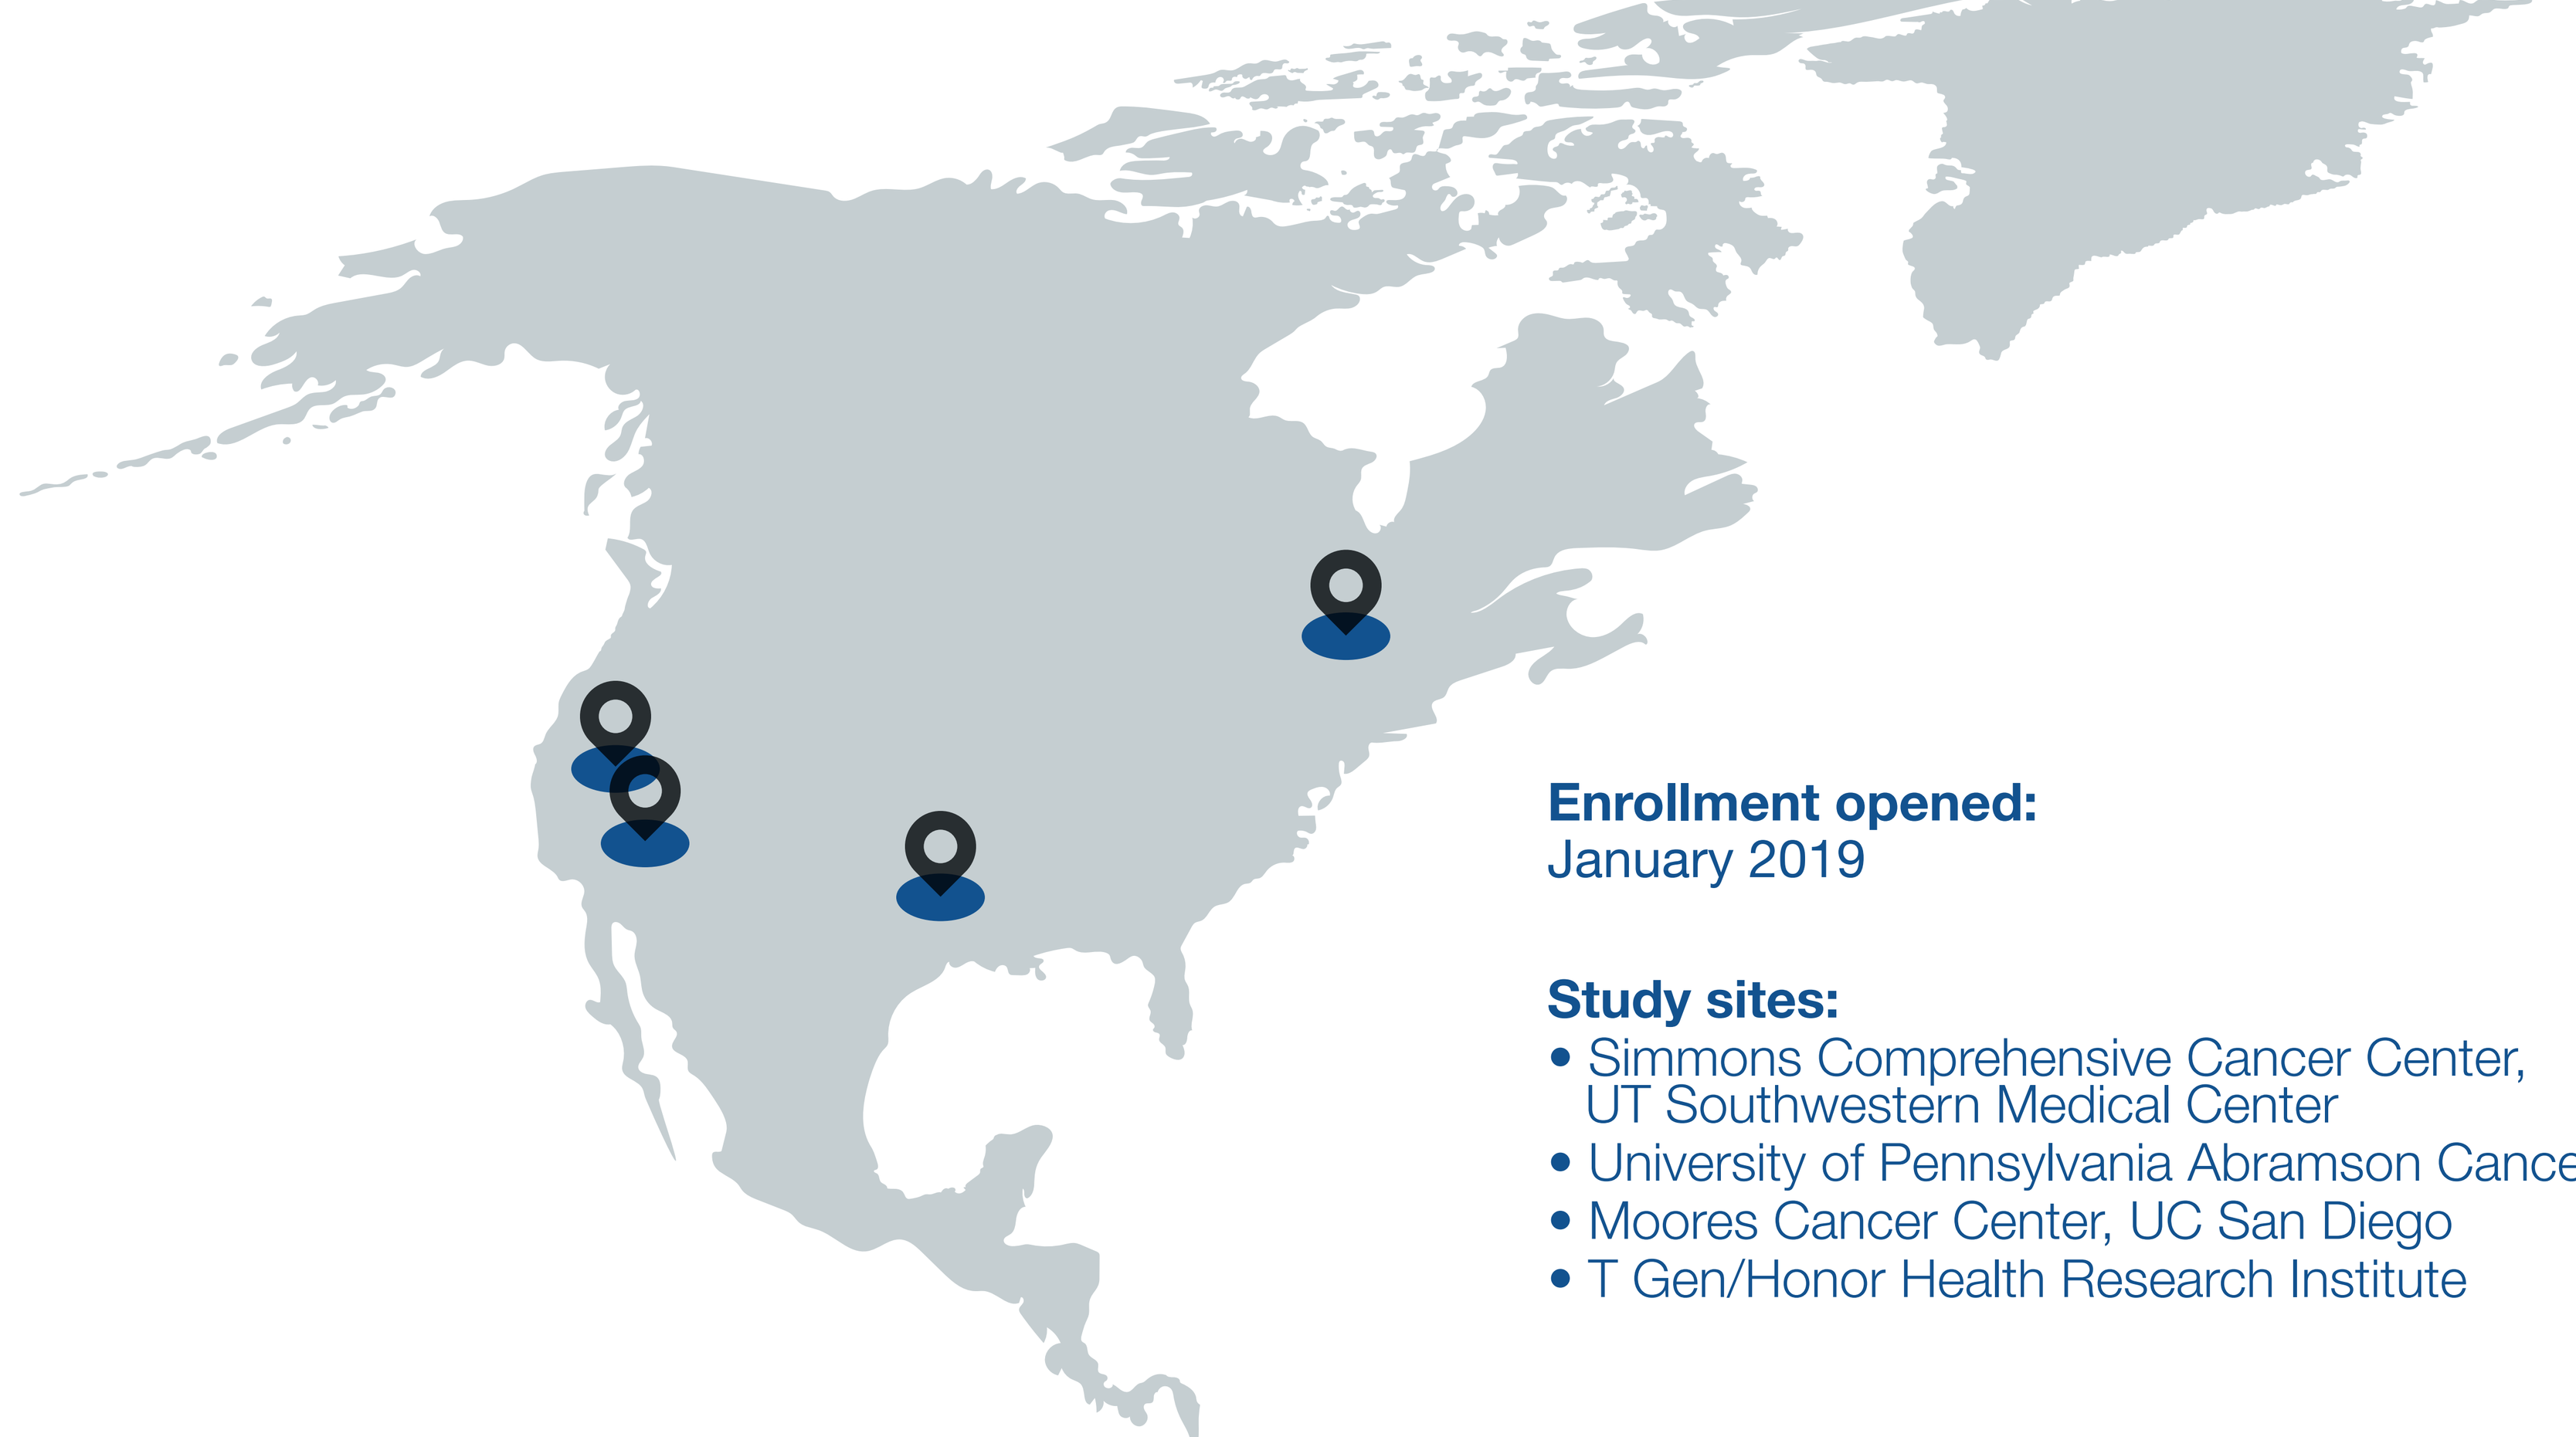

Enrollment opened:
January 2019

Study sites:

- Simmons Comprehensive Cancer Center, UT Southwestern Medical Center
- University of Pennsylvania Abramson Cancer Center
- Moores Cancer Center, UC San Diego
- T Gen/Honor Health Research Institute

Statistical Considerations

Safety run-in followed by a randomized phase 2 study

- To establish safety of the proposed combination, 3 -12 patients will be recruited in part 1/safety run-in based on modified 3+3 dose escalation rules.
- In part 2 of the study, 31 patients per arm will be enrolled in the randomization phase. The null hypothesis that the true complete response rate is 8.3% (historical control complete response rate) will be tested against a one-sided alternative hypothesis of the complete response rate of 23%. In the first stage,14 patients will be accrued in each arm. If there are 0 or 1 complete response in these 14 patients, the study arm will be stopped. Otherwise, 17 additional patients will be accrued for a total of 31 patients. The null hypothesis will be rejected if 5 or more complete responses are observed in 31 patients. This design yields an expected sample size of 19.5 patients with an early stopping probability of 67.4%.

Translational Analyses

To assess the effect of combination chemotherapy and bemcentinib on:

- AXL pathway activity in tumor tissue
- Changes in immune landscape including upregulation of immune cytokines, and immune cell infiltration into the tumor
- Apoptosis and decreased proliferation of tumor
- To identify predictive biomarkers of response

References

(1) Graham *et al.* The TAM family: phosphatidylserine sensing receptor tyrosine kinases gone awry in cancer. Nat Rev Cancer. 2014 Dec;14(12):769-85

(2) Ludwig *et al.* Small-Molecule Inhibition of Axl targets tumor immune suppression and enhances chemotherapy in pancreatic cancer. Cancer Res 2018;78:246-55

(3) Jameson *et al.* A phase Ib/II pilot trial with nab-paclitaxel plus gemcitabine plus cisplatin in patients with stage IV pancreatic cancer. Journal of Clinical Oncology 2017;35, no.4_suppl 341

Contact

Muhammad Shaalan Beg, MD MS
2201 Inwood Rd
Dallas 75235, TX
USA

muhammad.beg@utsouthwestern.edu
+1 214-645-4673
@ShaalanBeg

Clinical trial identifier
NCT03649321

This presentation is the intellectual property of the presenter. Contact muhammad.beg@utsouthwestern.edu for permission to reprint and/or distribute.

This poster was presented at ASCO Gastrointestinal Cancers Symposium 2019, 17-19 January, San Francisco, CA